Poster #127

5th International Workshop on HIV Drug Resistance and Treatment Strategies, June 4-8, 2001

Analysis of Clinically Relevant Breakpoints for Lopinavir/r in Multiple PI-Experienced, NNRTI-Naive Patients Using Virtual Phenotype

D Kempf[1], R Harrigan[2], M King[1], J Isaacson[1], S Brun[1], K Garren[1], A Hsu[1], B Larder[2], E Sun*[1]

[1]Abbott Laboratories, Abbott Park, IL; [2]Virco, Cambridge, UK

ABSTRACT

Background: The virologic response to therapy with lopinavir/ritonavir (lopinavir/r) plus efavirenz and NRTIs in multiple PI-experienced, NNRTI-naïve patients has been shown to be associated with baseline lopinavir susceptibility and the number of PI resistance mutations (Study M98-957). In this analysis, the response at Week 48 in this study with respect to the baseline lopinavir virtual phenotype was examined.

Methods: Baseline protease (Pr) genotypes (n=56), determined by population sequencing, were used to calculate prototype virtual phenotypes for lopinavir using a database of >1000 isolates and a neural network approach. Baseline phenotypes were previously determined using the PhenoSense™ method. Statistical analysis was performed using Fisher's Exact Test.

Results: The median baseline lopinavir virtual phenotype was 11.4-fold (range 0.9- to 50.2-fold). The median ratio of VirtualPhenotype™/PhenoSense™ phenotype was 1.2 (range 0.29 to 9.0). The Pearson correlation coefficient for $\log_{10}$ virtual phenotype vs. $\log_{10}$ phenotype was 0.87. At Week 48, HIV RNA <400 copies/mL was observed in 23/25 (92%) and 15/25 (60%) subjects with lopinavir virtual phenotypes of <15-fold and ≥15-fold, respectively (p=0.018). HIV RNA <50 copies/mL was observed in 20/25 (80%) and 13/25 (52%) of the above groups of subjects, respectively (p=0.072). An exploration of various potential virtual phenotype breakpoints (5-fold, 10-fold, 15-fold, 20-fold and 25-fold) suggested that 10- or 15-fold breakpoints were better predictors than other potential breakpoints. Virologic response was also associated with virtual inhibitory quotient (vIQ): at Week 48, HIV RNA <400 copies/mL was observed in 21/21 (100%) and 17/29 (59%) of subjects with vIQ >7 and <7, respectively (p<0.001).

Conclusions: Lopinavir virtual phenotype, as derived from baseline genotype, correlates with clinical response in PI-experienced patients. A breakpoint in the range of 10- to 15-fold is similar to that observed using actual lopinavir phenotype.

INTRODUCTION

Lopinavir/ritonavir (LPV/r, Kaletra™) is a new protease inhibitor (PI) that has shown potent activity in both treatment-naïve and PI-experienced patients. In Study M97-720, treatment of 100 ARV-naïve patients with LPV/r plus d4T/3TC produced a virologic response to HIV RNA <400 copies/mL in 99% of subjects on treatment (80% ITT missing=failure), respectively, after 108 weeks (Stryker, 2000). In Study M98-957, treatment of 57 multiple PI-experienced, NNRTI-naïve patients with LPV/r plus efavirenz (EFV) and nucleoside reverse transcriptase inhibitors (NRTIs) of choice produced a decline in HIV RNA to <400 copies/mL in 80% (on treatment) and 65% (ITT missing=failure) of subjects after 48 weeks (Rockstroh, 2000). The phenotypic and genotypic breakpoints for LPV/r in Study M98-957 have been determined. Thus, response was higher in those subjects whose baseline susceptibility to LPV *in vitro* was attenuated by <10-fold, compared to those with ≥10-fold reduced susceptibility (Kempf, 2000a). Similarly, response was higher in subjects whose baseline isolates contained 5 or fewer of the 11 mutations associated with reduced susceptibility to LPV (LPV mutation score ≤5), than in those with LPV mutation score of ≥6 (Kempf, 2000a, Kempf, 2000b).

The virtual phenotype (VirtualPhenotype™) is a rapid, automated method for quantitative prediction of HIV-1 phenotypic susceptibility from a genetic sequence using matched samples from a relational database (Larder, 2000). With new agents such as LPV, the development of a virtual phenotype database requires the determination of phenotypic susceptibility of a large panel of clinical isolates. In addition, where clinical data are available, the definition of breakpoints for virtual phenotype is important for proper interpretation of these assays.

Virologic response in Study M98-957 has also been evaluated with respect to the LPV inhibitory quotient (IQ). Defined as the ratio of C_{trough} to IC_{50} (adjusted for human serum binding), the IQ provides an estimate of *in vivo* potency that reflects both *in vitro* virologic activity and plasma levels in HIV-infected patients. In Study M98-957, virologic response at Week 24 was highest in those subjects with LPV IQ >15, intermediate in those subjects with LPV IQ between 4 and 15, and lowest in subjects with LPV IQ <4 (Hsu, 2000). IQ values based on a virtual phenotype (virtual IQ) have been shown to be predictive of virologic response to ritonavir intensification of therapy in patients with detectable viral load while receiving indinavir (Kempf, 2001).

In this study, we have determined prototype virtual phenotypes for the baseline viruses from Study M98-957, and assessed the virologic response at Week 48 with respect to both virtual phenotype and virtual IQ. The results provide an estimate of the appropriate virtual phenotype breakpoint for LPV/r.

METHODS

- Plasma HIV RNA was quantified using Roche Amplicor HIV-1 Monitor™* (lower limit of quantitation [LLQ] 400 copies/mL) or the Roche Ultrasensitive assay (LLQ 50 copies/mL)
- Baseline phenotype was measured by the PhenoSense™ method, as previously reported (Kempf, 2000b). Baseline genotype was determined by population sequencing.
- Virtual phenotype was estimated for LPV using a database of >1000 isolates for which protease genotype and susceptibility to LPV had been determined, and a neural network approach. (Larder, 2000).
- C_{trough} was determined at steady state (Week 5). Virtual inhibitory quotient values for each patient were calculated by the following equation:

$$\text{Virtual IQ} = \frac{\text{observed } C_{trough}}{\text{virtual phenotype} \times IC_{50}\ (\text{wt HIV, 50\% human serum*})}$$

- The serum-adjusted IC_{50} value for LPV (0.059 µg/mL) represents the mean IC_{50} for three wild-type laboratory strains (IIIB, HXB2 and pNL4-3) in the presence of 50% human serum plus 10% fetal calf serum (Molla, 1998). The free fraction of LPV in 50% human serum plus 10% fetal calf serum has been shown to approximate the free fraction in 100% human serum (Hsu, 2001).
- Virologic response was defined by plasma HIV RNA ≤400 or ≤50 copies/mL at Week 48 using a Dropouts as Censored definition (DeGruttola, 2000). Statistical analysis was by Fisher's Exact Test.

RESULTS

Characterization of Baseline Isolates

- Phenotype/genotype was obtained on 56/57 baseline viruses (Table 1).
- The median number of prior PIs, prior NRTIs, and prior antiretroviral agents was 3, 4 and 7.5 respectively.
- The prevalence of multiple baseline mutations associated with cross resistance to the PI class was high (Figure 1). The majority (79%) of isolates contained one or more primary mutations at positions 82, 84 and 90, as well as secondary mutations at positions 10 and/or 71.
- With four exceptions, the baseline isolates were uniformly sensitive to EFV.

Table 1. Characterization of Study M98-957 Baseline Isolates (n=56)

Antiviral Drug	Previous Therapy (%)	Isolates with >4-Fold Reduced Susceptibility (%)
LPV	0	55
IDV	86	68
RTV	77	73
SQV	71	59
NFV	57	77
APV	0	30
d4T	93	11
ZDV	93	71
3TC	91	70
ddI	80	2
ddC	46	4
ABC	18	55
EFV	0	4

Figure 1. Prevalence of Baseline Protease Mutations Associated with PI Resistance

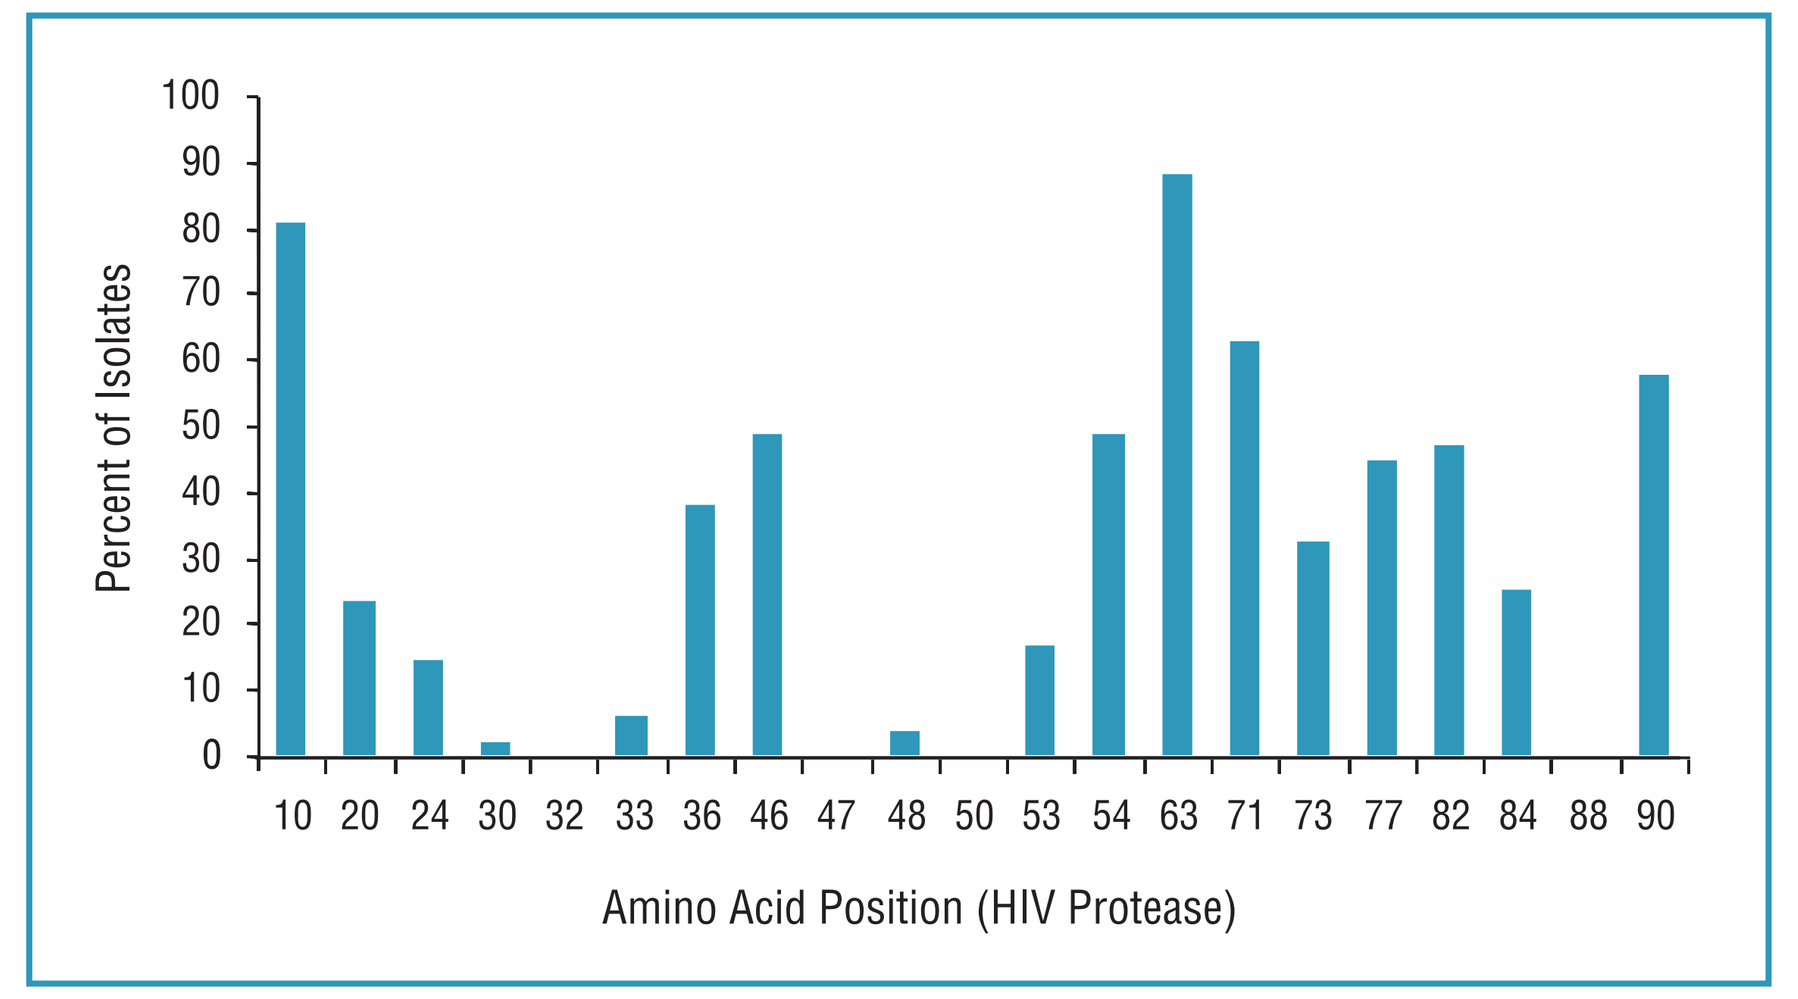
Percent of Isolates
100
90
80
70
60
50
40
30
20
10
0
10 20 24 30 32 33 36 46 47 48 50 53 54 63 71 73 77 82 84 88 90
Amino Acid Position (HIV Protease)

- The LPV phenotype (PhenoSense) of the baseline isolates ranged from 0.5- to 96-fold. LPV virtual phenotype (*virtual*Phenotype) for the same isolates ranged from 0.9- to 50-fold. The mean phenotype and virtual phenotype were 16.2-fold and 15.6-fold, respectively. Median phenotype and virtual phenotype were 5.2-fold and 11.4-fold, respectively.
- The correlation of phenotype and virtual phenotype across a *ca.* 2-log range was relatively high (Figure 2), with a Pearson correlation coefficient for $\log_{10}$ virtual phenotype vs. $\log_{10}$ phenotype of 0.87. The median ratio of virtual phenotype/phenotype for the group of individual baseline isolates was 1.16.
- Baseline categorical descriptors of virtual phenotype are provided in Figure 3. The prevalence of isolates with >15-fold virtual phenotype was 26/56 (46%).

Figure 2. Correlation of Phenotype and Virtual Phenotype

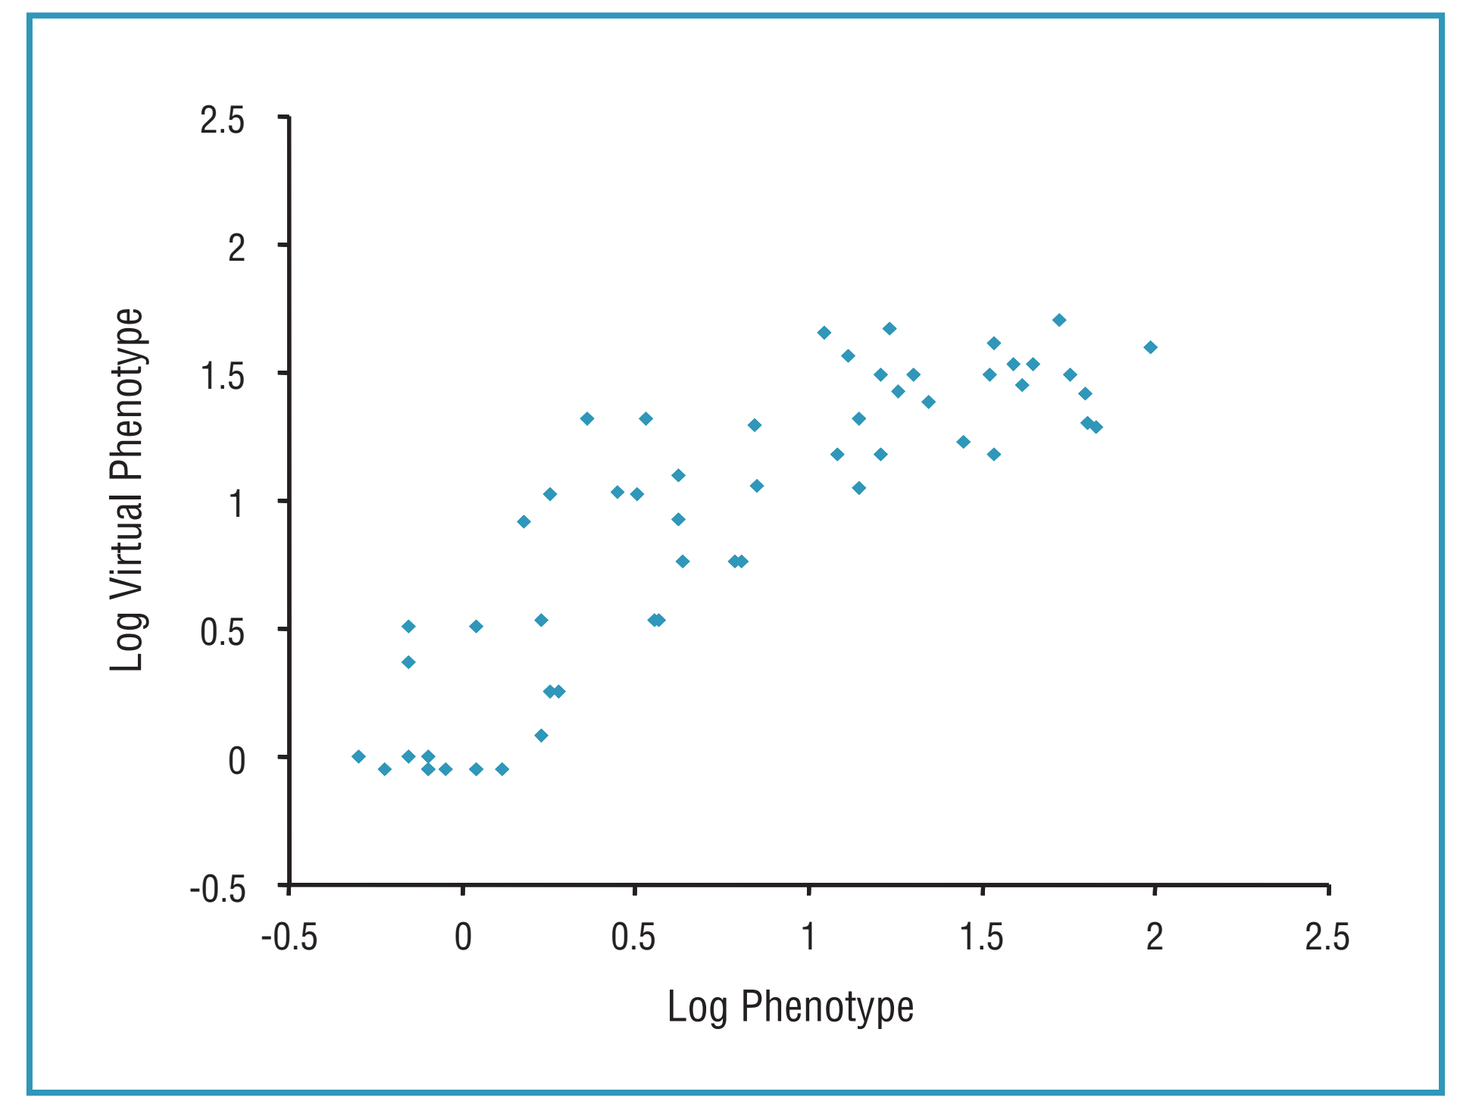
Log Virtual Phenotype
2.5
2
1.5
1
0.5
0
-0.5
-0.5 0 0.5 1 1.5 2 2.5
Log Phenotype

Figure 3. Baseline Virtual Phenotype for Study M98-957

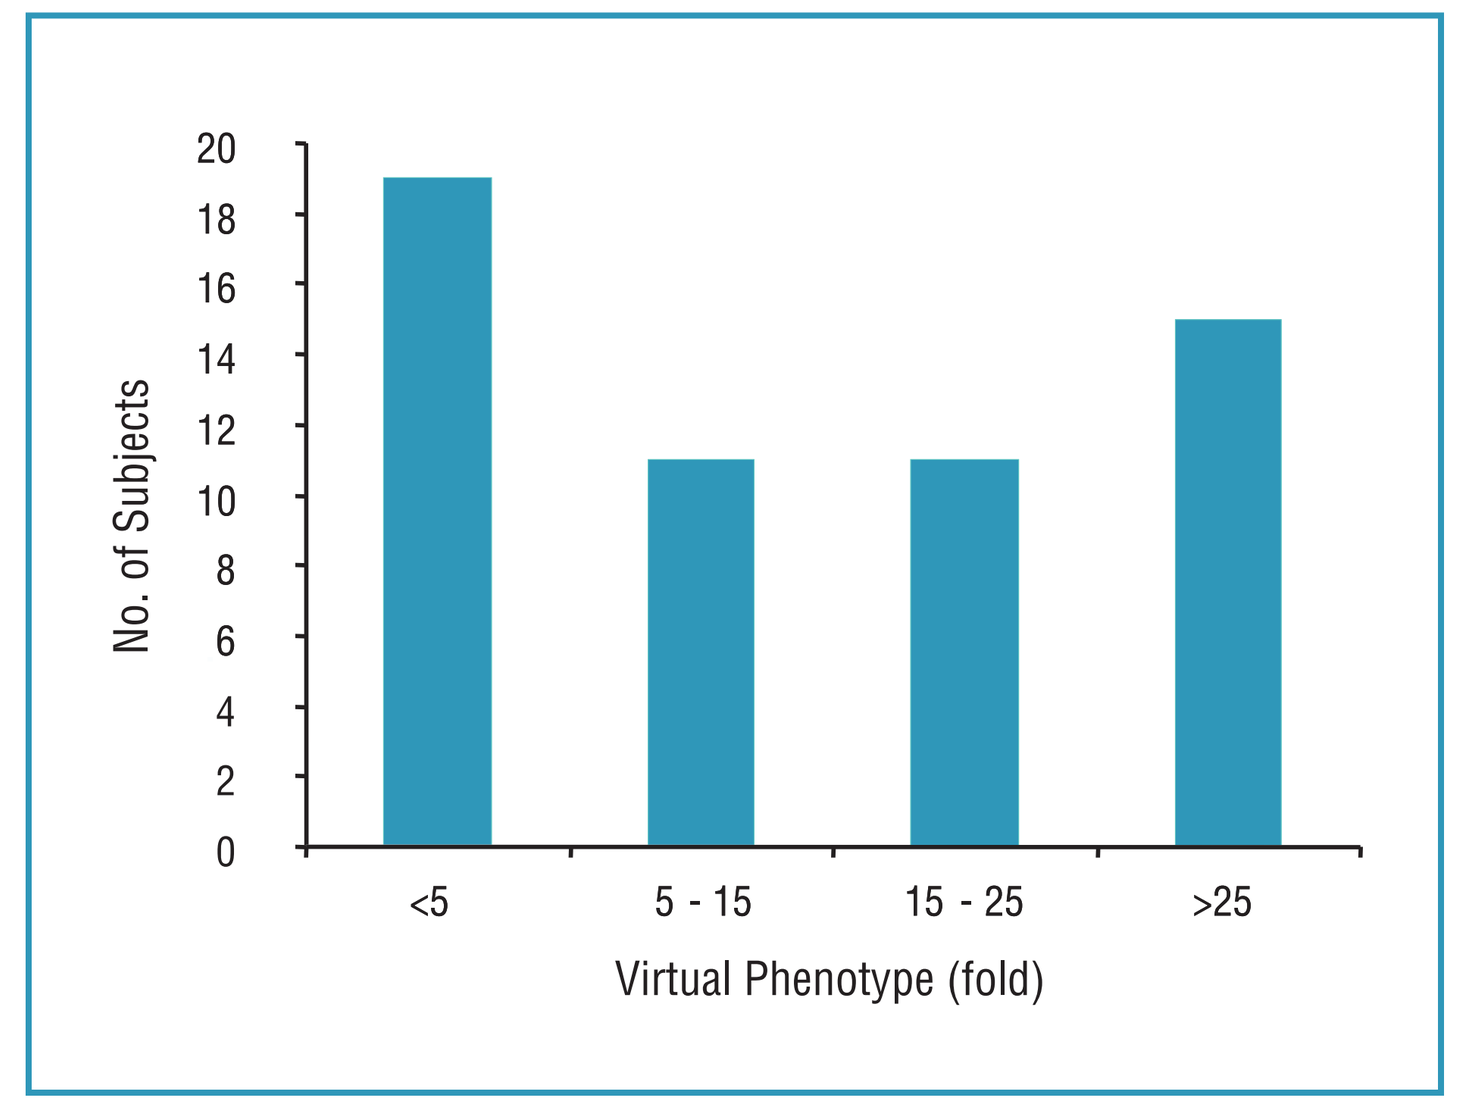
No. of Subjects
20
18
16
14
12
10
8
6
4
2
0
<5
5 - 15
15 - 25
>25
Virtual Phenotype (fold)

Analysis of Virologic Response with Respect to Baseline Virtual Phenotype

- Virologic response at both 400 and 50 copies/mL cutoff values was evaluated using a Dropouts as Censored definition. The disposition of the 57 subjects at Weeks 24 and 48 is provided in Table 2.

Table 2. Disposition of Subjects in Study M98-957 at Weeks 24 and 48

Disposition	Number of Subjects Week 24	Number of Subjects Week 48	Response Score
HIV RNA ≤50 copies/mL	35	33	Response
HIV RNA >50 and ≤400 copies/mL	7	5	Response*
HIV RNA >400 copies/mL	7	8	Failure
Discontinued therapy between Week 8 and Week 24 with HIV RNA >400 copies/mL	3	3	Failure
Discontinued therapy between Week 24 and Week 48 with HIV RNA >400 copies/mL	n/a	1	Failure
Discontinued therapy between Week 24 and Week 48 with HIV RNA ≤400 copies/mL	n/a	1	Censored
Drug interruption Weeks 32-48	n/a	1	Censored
Discontinued therapy prior to Week 8	4	4	Censored
Baseline genotype and phenotype not available	1	1	Censored
Total	57	57	

*Response at 400 copies/mL level; failure at 50 copies/mL level.

- Virologic response (<400 copies/mL) was statistically significantly higher in subjects with baseline virtual phenotype <15-fold than in the subjects whose isolates displayed a virtual phenotype of >15-fold (p=0.018 for Week 48 comparison). Longitudinal response with respect to these categorical descriptors of virtual phenotype is provided in Figure 4.
- Virologic response to <50 copies/mL was also associated with baseline virtual phenotype (Figure 5).

Figure 4. Virologic Response (<400 copies/mL) with Response to Baseline Virtual Phenotype

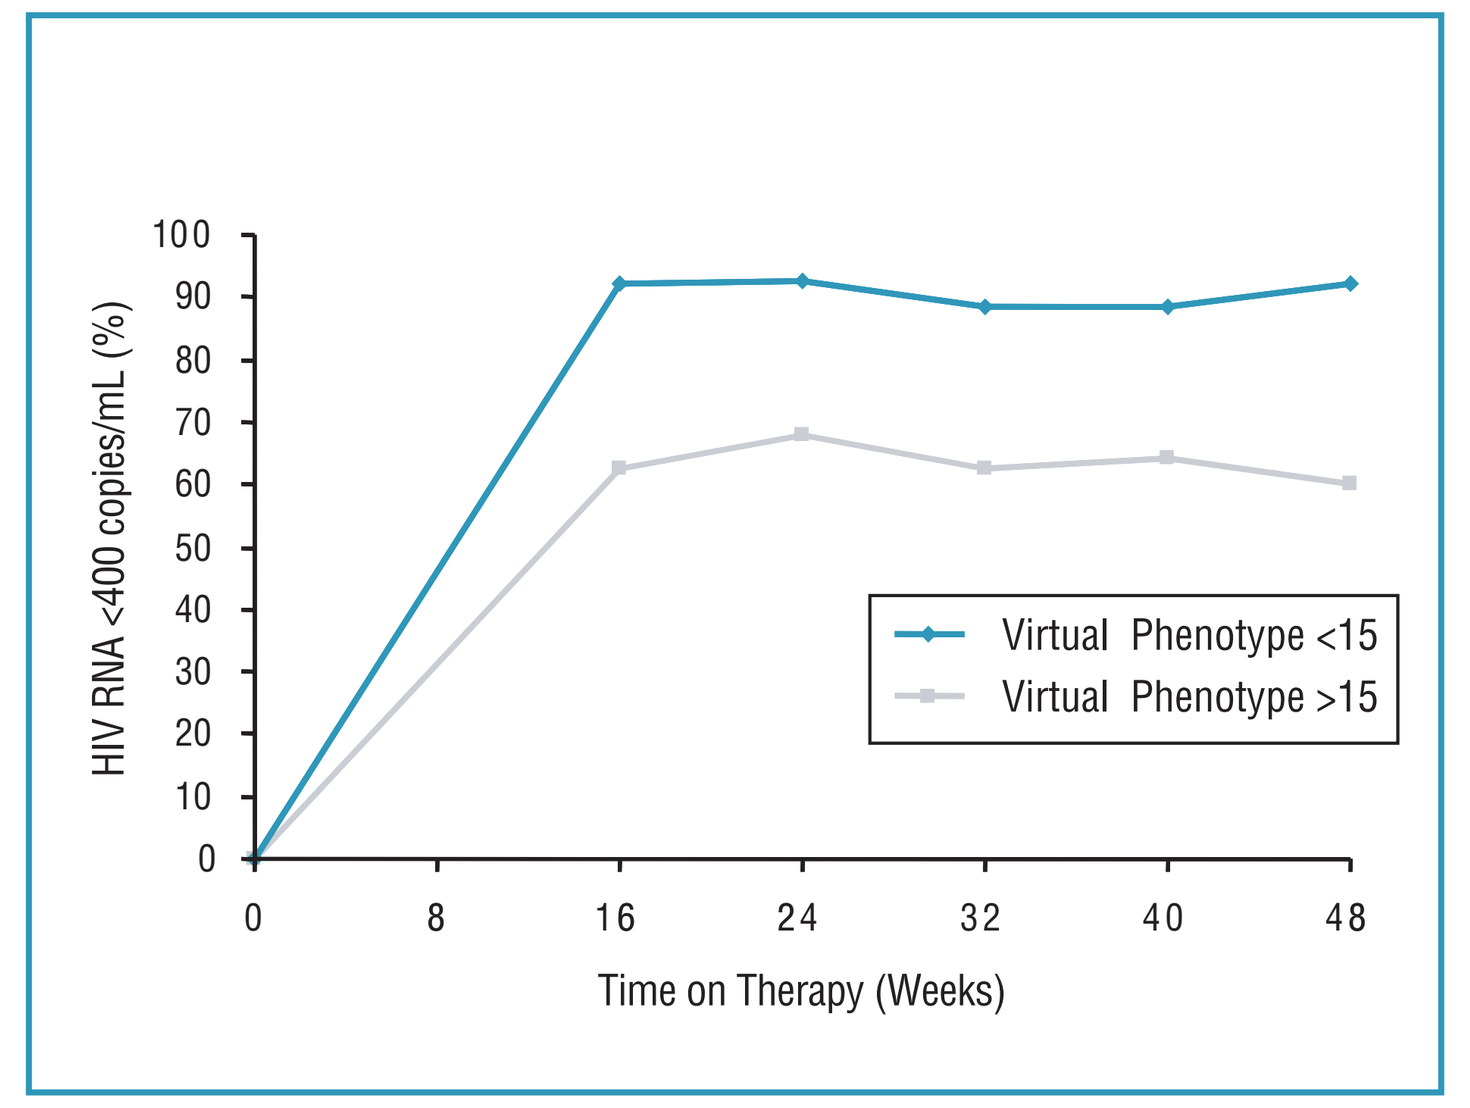
HIV RNA <400 copies/mL (%)
100
90
80
70
60
50
40
30
20
10
0
0 8 16 24 32 40 48
Time on Therapy (Weeks)
Virtual Phenotype <15
Virtual Phenotype >15

Figure 5. Virologic Response (Week 48) with Respect to Baseline Virtual Phenotype

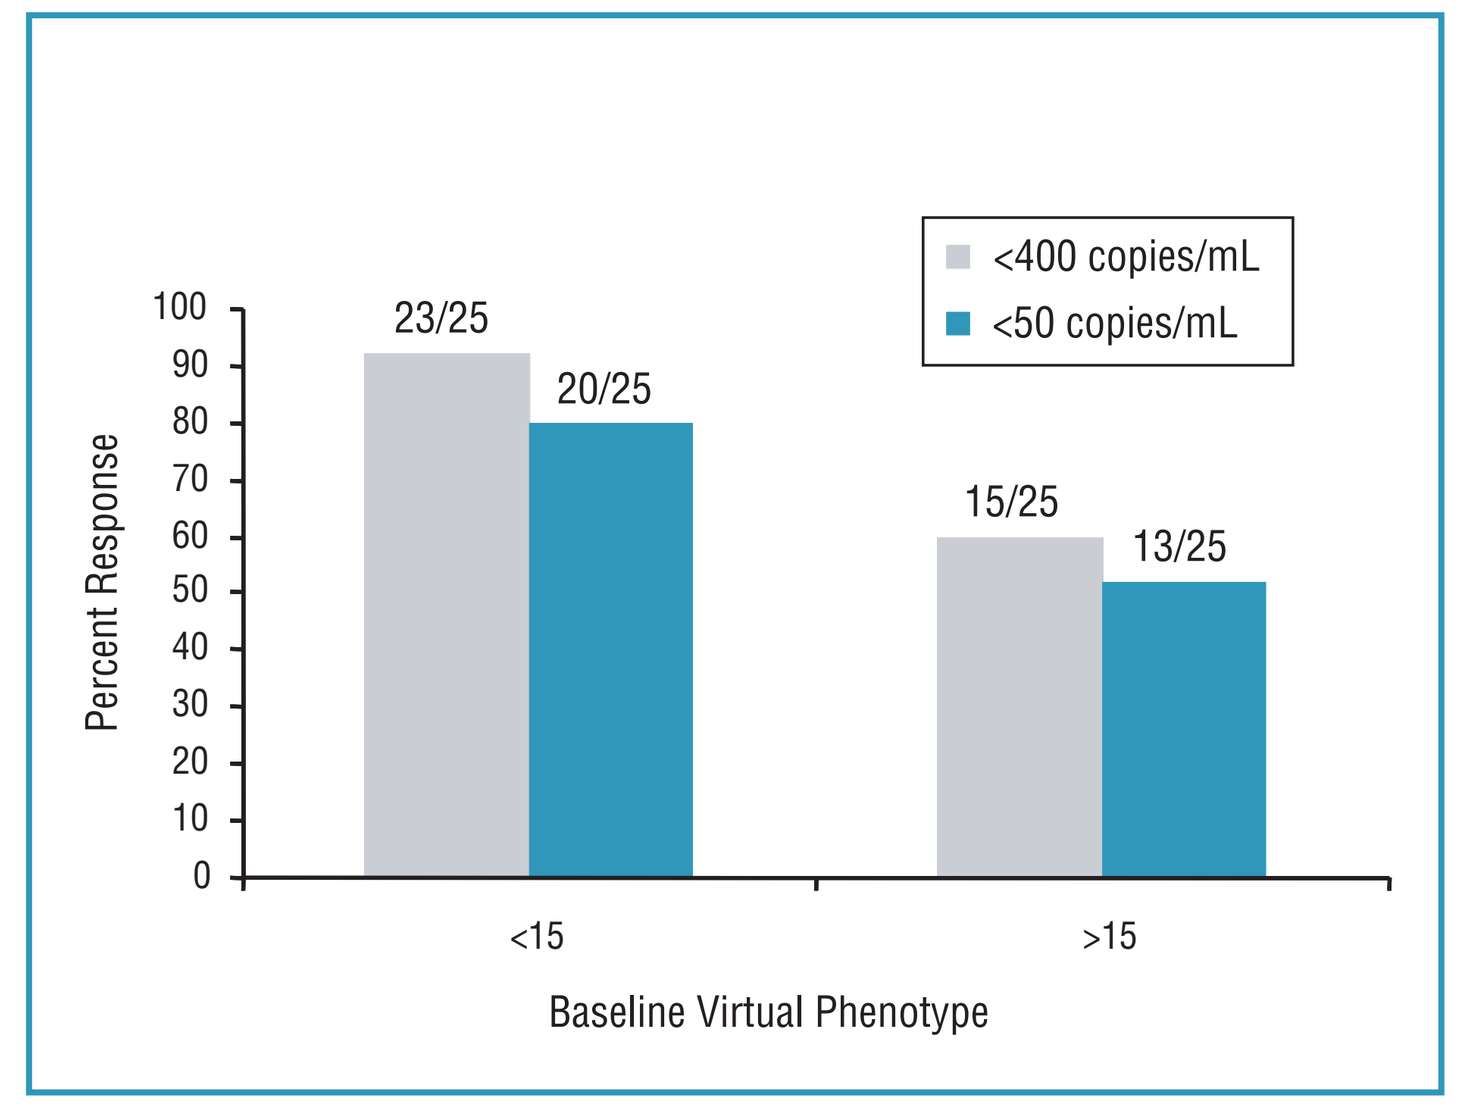
Percent Response
100
90
80
70
60
50
40
30
20
10
0
23/25
20/25
15/25
13/25
<400 copies/mL
<50 copies/mL
<15
>15
Baseline Virtual Phenotype

Analysis of the Virtual Phenotypic Breakpoint for LPV/r

- Potential virtual phenotypic breakpoints were examined for virologic response at Week 48 (Tables 3 and 4). Virtual phenotype breakpoints of 15-fold and 10-fold provided the most robust differentiation between the likelihood of virologic response and failure.

Table 3. Virologic Response (<400 copies/mL, Week 48) with Respect to Potential Virtual Phenotype Breakpoints

Potential Breakpoint	Response with Respect to Breakpoint: Below	Response with Respect to Breakpoint: Above	p-value
5-fold	16/17 (94%)	22/33 (67%)	0.039
10-fold	19/20 (95%)	19/30 (63%)	0.016
15-fold	23/25 (92%)	15/25 (60%)	0.018
20-fold	25/31 (81%)	13/19 (68%)	0.496
25-fold	29/35 (83%)	9/15 (60%)	0.416

Table 4. Virologic Response (<50 copies/mL, Week 48) with Respect to Potential Virtual Phenotype Breakpoints

Potential Breakpoint	Response with Respect to Breakpoint: Below	Response with Respect to Breakpoint: Above	p-value
5-fold	13/17 (76%)	20/33 (61%)	0.351
10-fold	16/20 (80%)	17/30 (57%)	0.129
15-fold	20/25 (80%)	13/25 (52%)	0.072
20-fold	21/31 (68%)	12/19 (63%)	0.767
25-fold	25/35 (71%)	8/15 (53%)	0.329

Analysis of Virologic Response with Respect to Virtual Inhibitory Quotient

- Baseline categorical descriptors of LPV virtual IQ are provided in Figure 6. The number of patients with LPV virtual IQ >10 and <10 were 21/52 (40%) and 31/52 (60%), respectively.

Figure 6. Baseline Virtual IQ for Study M98-957

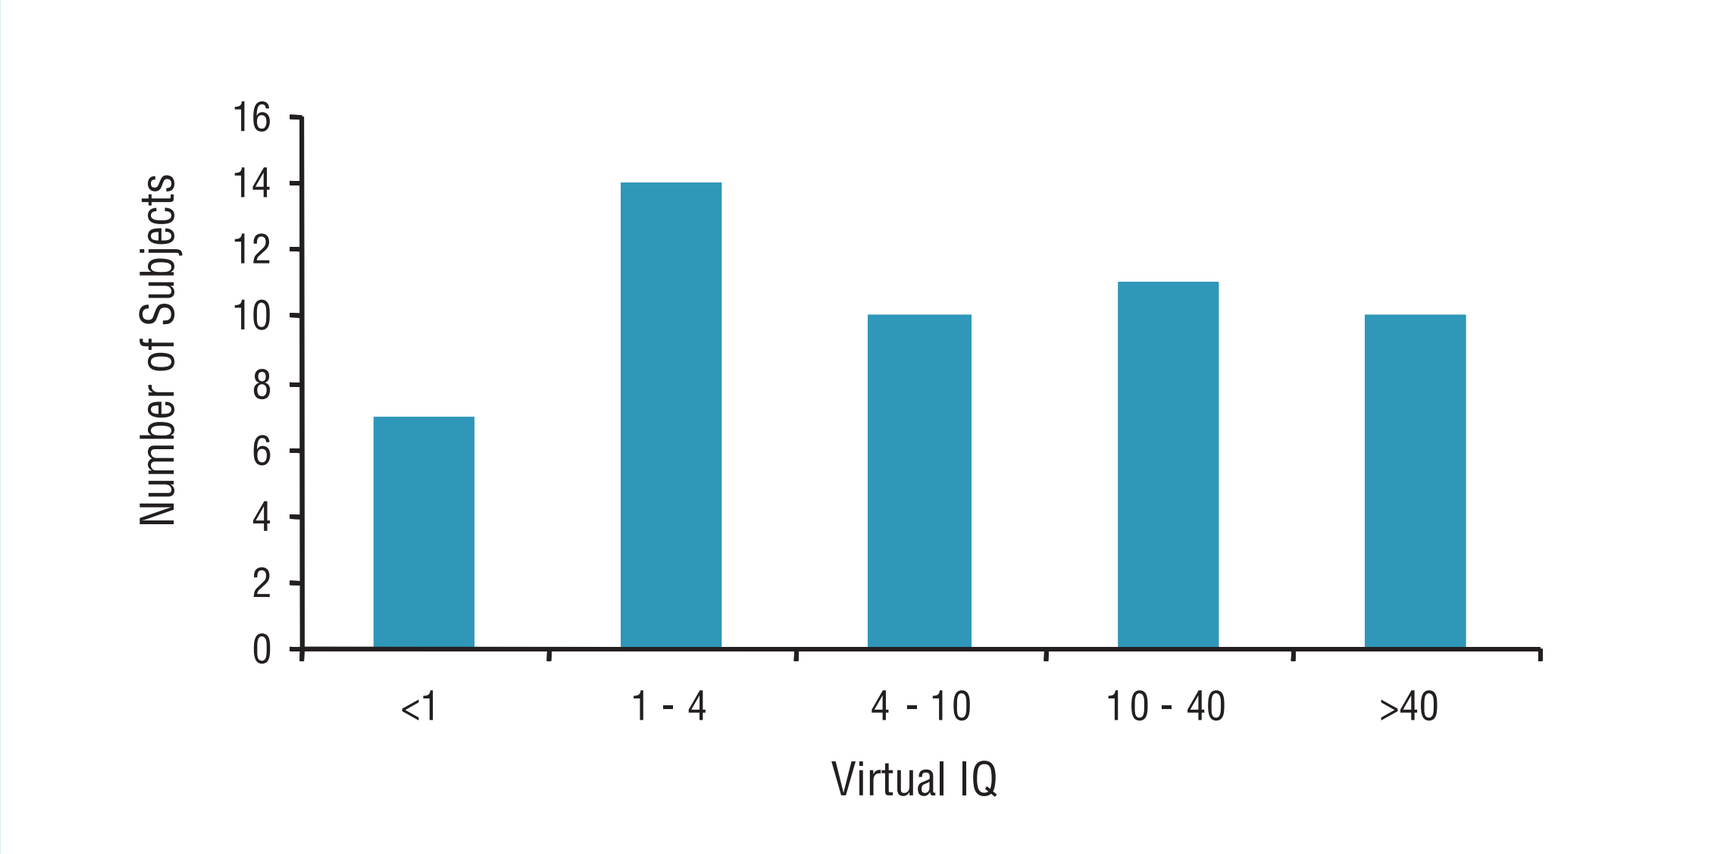
Number of Subjects
16
14
12
10
8
6
4
2
0
<1
1 - 4
4 - 10
10 - 40
>40
Virtual IQ

- Longitudinal response with respect to baseline virtual phenotype is provided in Figure 7. At Week 48, HIV RNA <400 copies/mL was statistically significantly higher in subjects with LPV virtual IQ >10 than in subjects with LPV virtual IQ <10 (p=0.001, Figure 8). Virologic response to <50 copies/mL was also higher for patients with virtual IQ >10 (16/20, 80%) vs. virtual IQ <10 (17/30, 57%), but the difference was not statistically significant (p=0.129).

Figure 7. Longitudinal Response (<400 copies/mL) with Respect to Baseline Virtual IQ

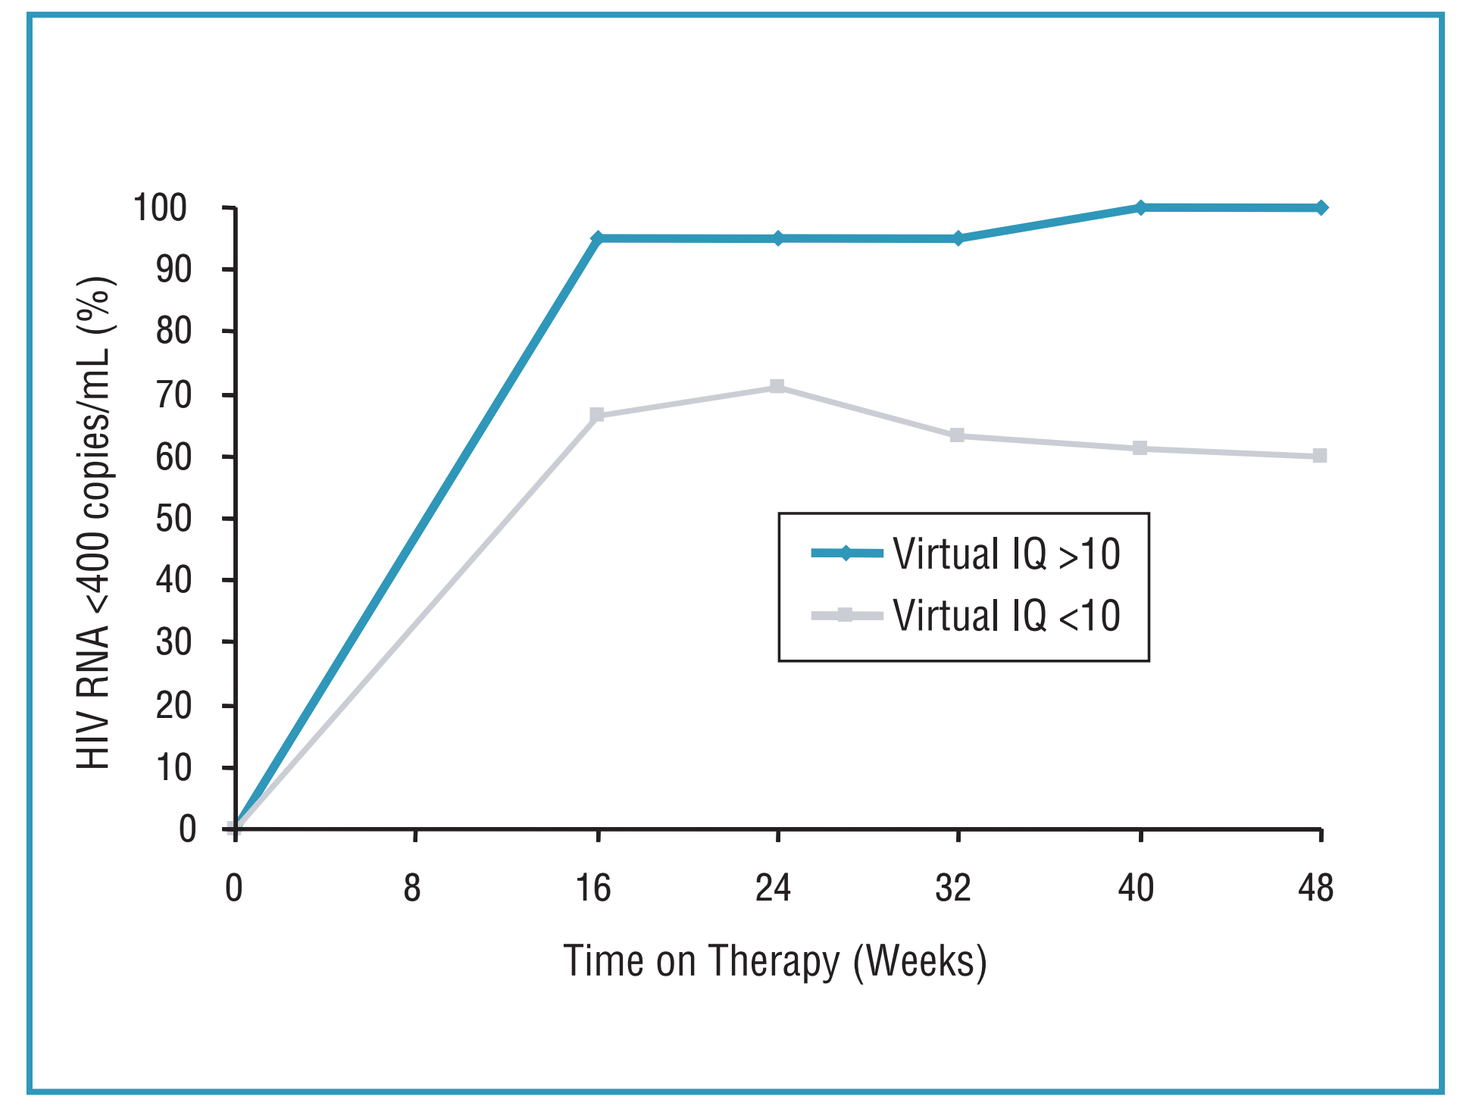
HIV RNA <400 copies/mL (%)
100
90
80
70
60
50
40
30
20
10
0
0 8 16 24 32 40 48
Time on Therapy (Weeks)
Virtual IQ >10
Virtual IQ <10

Figure 8. Virologic Response (Week 48) with Respect to Baseline Virtual IQ

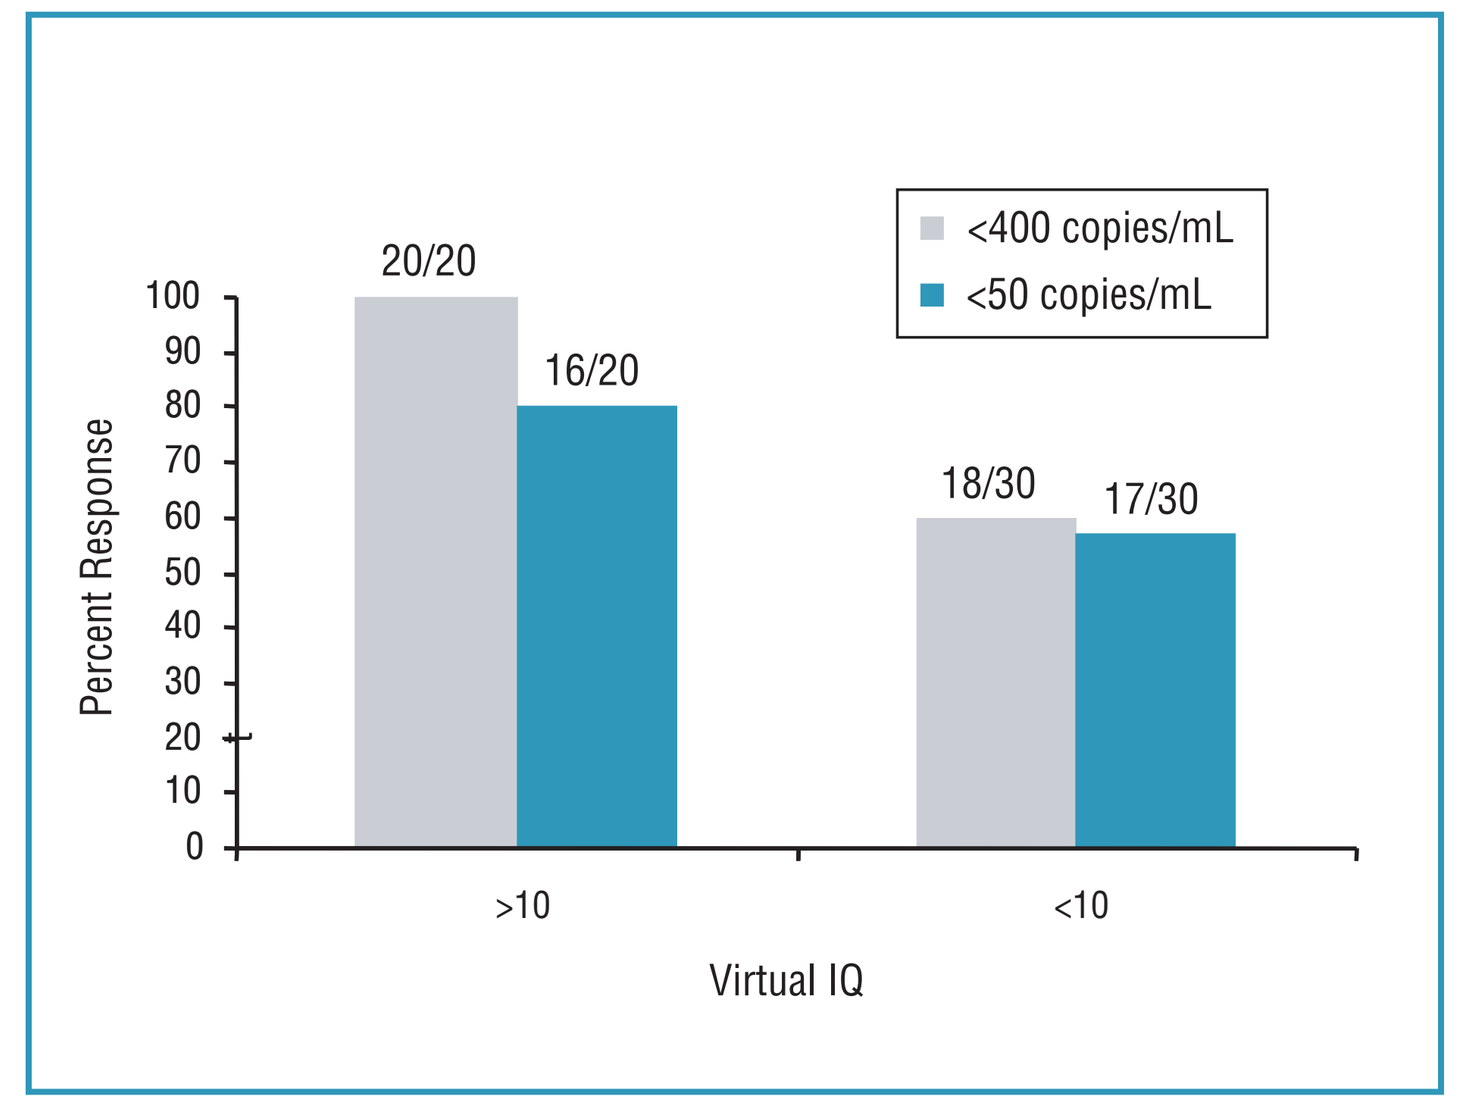
Percent Response
100
90
80
70
60
50
40
30
20
10
0
20/20
16/20
18/30
17/30
<400 copies/mL
<50 copies/mL
>10
<10
Virtual IQ

CONCLUSIONS

- LPV virtual phenotype, as derived from baseline genotype, correlates with virologic response in PI-experienced patients at both 400 and 50 copies/mL cutoffs.
- A breakpoint in the range of 10- to 15-fold for LPV/r is similar to that observed using actual LPV phenotype.
- The LPV virtual IQ, derived from the virtual phenotype and LPV trough levels, is also predictive of virologic response.

REFERENCES

DeGruttola, V. et al., 2000, *Antiviral Therapy* **5**, 41.
Hsu, A. et al., 2001, 8th Conference on Retroviruses and Opportunistic Infections, Chicago, IL, Abstract 753.
Kempf, D. et al., 2000a, 4th International Workshop on HIV Drug Resistance & Treatment Strategies, Sitges, Spain, Abstract 89.
Kempf, D. et al., 2000b, 4th International Workshop on HIV Drug Resistance & Treatment Strategies, Sitges, Spain, Abstract 38.
Kempf, D. et al., 2001, 8th Conference on Retroviruses and Opportunistic Infections, Chicago, IL, Abstract 523.
Larder, B. A. et al., 2000, 4th International Workshop on HIV Drug Resistance & Treatment Strategies, Sitges, Spain, Abstract 62.
Molla, A. et al., 1998, *Virology* **250,** 255.
Rockstroh, J. et al. 2000, 5th International Congress on Drug Therapy in HIV Infection, Glasgow, UK, Abstract P43.
Stryker, R. et al. 2000, 5th International Congress on Drug Therapy in HIV Infection, Glasgow, UK, Abstract P46.

E011680F