Poster #61

5th International Workshop on HIV Drug Resistance and Treatment Strategies, June 4-8, 2001

Analysis of Mutational Patterns Associated with Response to Lopinavir/ritonavir in Multiple PI-Experienced Patients

D Kempf, M King, J Isaacson, R Rode, S Brun, E Sun; Abbott Laboratories, Abbott Park, IL USA

ABSTRACT

Background: In multiple PI-experienced, NNRTI-naïve subjects, virologic response to lopinavir/ritonavir (lopinavir/r) plus efavirenz and NRTIs was significantly higher at Week 48 in those subjects with a baseline lopinavir mutation score of ≤5 (21/23, 91%) than in those with a baseline lopinavir mutation score of ≥6 (17/27, 63%; p=0.024). In order to investigate mutational patterns associated with virologic failure, baseline characteristics of subjects with ≥6 baseline mutations were examined.

Methods: Viral phenotype was determined using a single cycle recombinant assay. Genotype was determined by population sequencing. Virologic response was defined as plasma HIV RNA <400 copies/mL (dropouts as censored). The Wilcoxon rank sum test was used for univariate analysis of baseline parameters.

Results: All (10/10) of the baseline isolates from subjects failing therapy with lopinavir mutation score ≥6 had mutations at positions 82, 54 and 10 plus a median of 4 additional PI mutations. This pattern was associated with lower response (p=0.0017). However, an equal number of subjects who experienced response through Week 48 also had mutations at positions 82, 54 and 10 plus a median of 4 additional PI mutations. Within this group of 20 subjects, baseline fold susceptibility to lopinavir (median 51- and 16-fold, respectively) was a univariate predictor of response (p=0.014), suggesting that in viruses carrying multiple, but similar numbers of mutations, phenotype provides additional information above genotype. In the absence of a mutation at position 82, virologic response at Week 48 was observed in 14/15 subjects with a baseline L90M mutation (6/6 with baseline I84V/L90M).

Conclusions: A pattern containing PI mutations at positions 82, 54, 10 plus multiple other mutations at baseline is associated with increased risk of failure of lopinavir/r therapy in multiple PI-experienced subjects. However, with complicated baseline genotypes of ≥6 mutations, viral phenotype may provide additional predictive power over genotype. In contrast to the above pattern containing a mutation at position 82, mutations at positions 84 and/or 90 in the context of other mutations are not predictive of failure.

BACKGROUND

Lopinavir/ritonavir (LPV/r, Kaletra™) is a conformulation of lopinavir, an HIV protease inhibitor, and ritonavir, which inhibits LPV metabolism by cytochrome P450 3A, providing increased plasma levels of LPV. The efficacy and safety of LPV/r-based regimens are currently being studied in HIV-infected patients, both antiretroviral-naive and PI-experienced. After 108 weeks, in an intent-to-treat (missing=failure) analysis, HIV RNA levels were <400 copies/mL for 80% of ARV-naive patients (on treatment: 99%) receiving LPV/r + d4T/3TC in Study M97-720 (Stryker, 2000).

Study M98-957 is an ongoing phase II, open-label, randomized trial of LPV/r in combination with efavirenz and NRTI(s) in multiple PI-experienced/ NNRTI-naive patients.

Figure 1. M98-957 Study Design

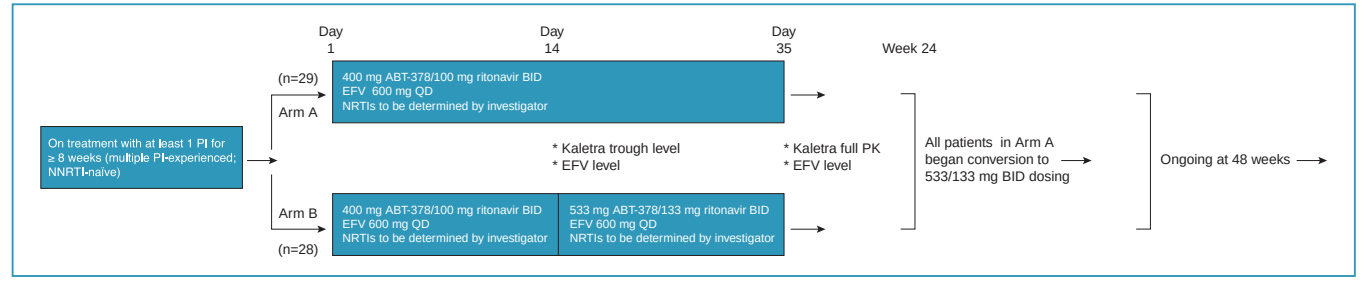
Day 1
Day 14
Day 35
Week 24
(n=29)
Arm A
400 mg ABT-378/100 mg ritonavir BID
EFV 600 mg QD
NRTIs to be determined by investigator
On treatment with at least 1 PI for ≥ 8 weeks (multiple PI-experienced; NNRTI-naive)
* Kaletra trough level
* EFV level
* Kaletra full PK
* EFV level
All patients in Arm A began conversion to 533/133 mg BID dosing
Ongoing at 48 weeks
Arm B
(n=28)
400 mg ABT-378/100 mg ritonavir BID
EFV 600 mg QD
NRTIs to be determined by investigator
533 mg ABT-378/133 mg ritonavir BID
EFV 600 mg QD
NRTIs to be determined by investigator

Viral isolates from 112 single and multiple PI-experienced patients have been analyzed to investigate the association of protease mutations with reduced phenotypic susceptibility to LPV (Kempf, 2000a). Eleven protease mutations have been identified that were associated with reduced *in vitro* susceptibility to LPV in viruses selected by other PIs: L10F, I, R, or V; K20M or R; L24I; M46I or L; F53L; I54L, T, or V; L63P; A71I, L, V, or T; V82A, F, or T; I84V; L90M. The LPV mutation score has been defined as the number of these mutations present in an isolate. Virologic response in Study M98-957 has been shown to be associated with the baseline LPV mutation score (Kempf, 2000b).

The average change in phenotypic susceptibility for incremental changes in the LPV mutation score was determined to be 1.74-fold per mutation (Kempf, 2000a). However, this analysis assumes an equal contribution of each mutation to reduced phenotypic susceptibility, which is unlikely. We therefore undertook an exploratory analysis of the relationship between protease mutational patterns and virologic response. Although the relatively small sample size of the study limits the power of this analysis to conclusively identify relationships, these observations provide insight into questions that may be addressed in larger studies (Calvez, Poster #82).

METHODS

- Viral genotype was determined at ViroLogic by population sequencing.
- Viral phenotype was determined using a single cycle recombinant assay (PhenoSense™).
- Virologic response was defined as plasma HIV RNA <400 copies/mL (or <50 copies/mL) at Week 48, using a dropouts as censored (DAC) analysis. In the DAC analysis, patients who discontinued the study prior to the timepoint of interest were censored if their final viral load was <400 copies/mL or were considered non-responders if the final viral load was above 400 copies/mL.
- The Data Analysis Plan (DAP) of the HIV Resistance Collaborative Working Group, which was created to provide a common analysis plan across studies to facilitate comparison of results from different studies of resistance, identifies the following PI mutations: L10F, I, R or V; K20M or R; L24I; D30N; V32I; L33F; M36I; M46I or L; I47V; G48V; I50V; I54L or V; A71V or T; G73S or A; V77I; V82A, F, S or T; I84V; N88D and L90M.
- For analyses involving the DAP mutations, the following PI mutations, present in the M98-957 baseline isolates and determined to be likely to be associated with resistance to the PI class by a review of public databases of wild-type and resistant HIV sequences, were added to the DAP list of PI mutations: I54T, A71I, A71L and G73T.
- Fisher's exact test was used to examine the association between virologic response and the presence of individual mutations or the LPV mutation score. A Wilcoxon rank-sum test was used to analyze the association of virologic response with certain baseline covariates, including phenotypic susceptibility to LPV and EFV, viral load, weight, and NRTI susceptibility.

RESULTS

Baseline LPV Mutation Score Is Associated with Virologic Response at Week 48

- As has been shown for earlier timepoints, virologic response (proportion <400 copies/mL, DAC analysis) at Week 48 was associated with baseline LPV mutation score (Figure 2).
- Similar patterns of response based on baseline LPV mutation score were observed using an ultrasensitive assay (<50 copies/mL) at Weeks 24 and 48 (Figure 3).

Figure 2. Virologic Response (Proportion <400 copies/mL, DAC) Through Week 48 Based on Baseline LPV Mutation Score

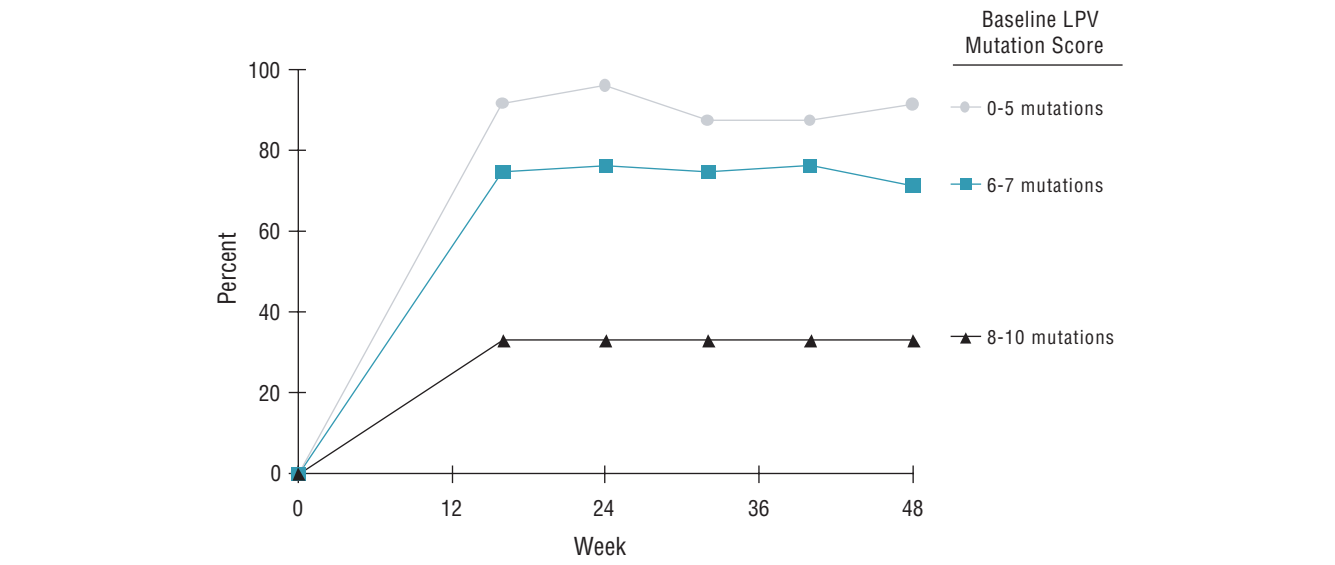
Baseline LPV Mutation Score
0-5 mutations
6-7 mutations
8-10 mutations
Percent
100
80
60
40
20
0
0
12
24
36
48
Week

Figure 3. Virologic Response (Proportion <50 copies/mL, DAC) Through Week 48 Based on Baseline LPV Mutation Score

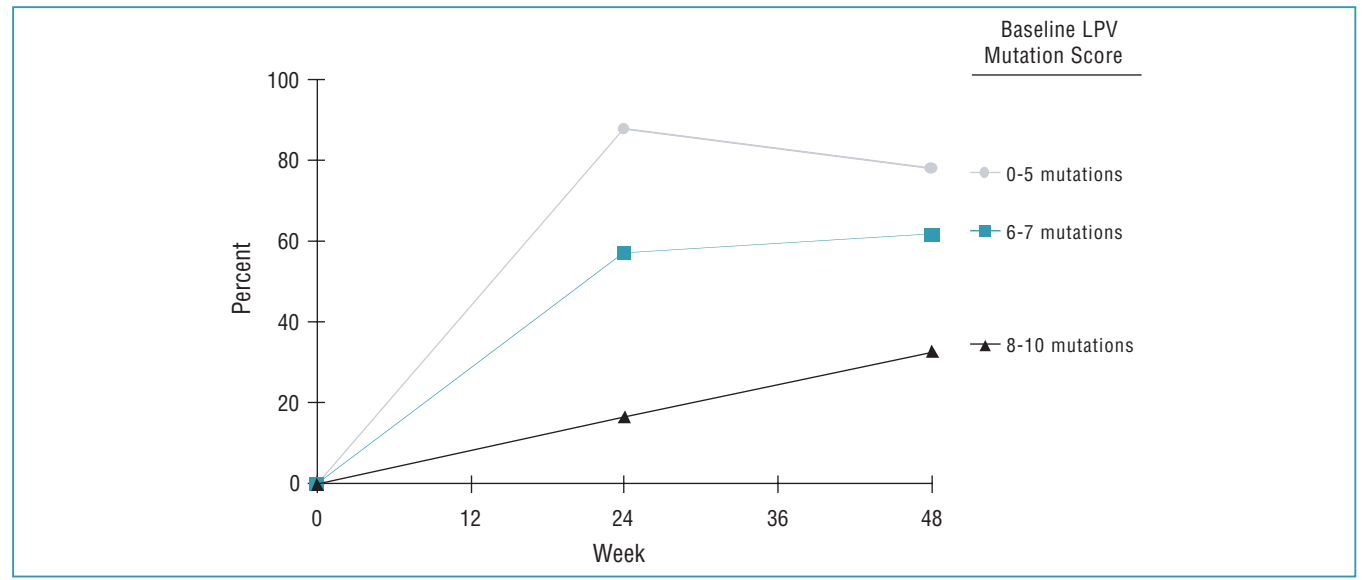
Baseline LPV Mutation Score
0-5 mutations
6-7 mutations
8-10 mutations
Percent
100
80
60
40
20
0
0
12
24
36
48
Week

Patterns of Mutations Associated with Week 48 Response

- The association of each individual mutation with Week 48 virologic response was analyzed using Fisher's exact test with a modified Bonferroni adjustment for multiple comparisons (p-values below 0.015 were considered statistically significant).
- Mutations at positions 54 and 82 were statistically significantly associated with Week 48 response, suggesting that they might be more important than other mutations in the LPV mutation score, although more than half the subjects with a mutation at position 54 or 82 experienced virologic response at Week 48, suggesting that the presence of these mutations alone was insufficient to describe resistance to LPV.
- Further review of the genotypic data for patients with baseline LPV mutation score ≥6 revealed that all 10 non-responders had mutations at positions 10, 54, and 82 along with at least 3 other PI mutations. This mutational pattern was highly associated with Week 48 virologic response: 28/30 (93%) patients without this pattern were Week 48 responders, compared to 10/20 (50%) patients with this pattern (p=0.007).
- However, there were as many responders as non-responders with this mutational pattern, suggesting that this pattern does not completely describe resistance to LPV.
- The mutations in the LPV mutation score demonstrated by the 20 patients with mutations at positions 82, 54, and 10 and a LPV mutation score of 6 or more are shown in Table 1, as are the other mutations included in the modified DAP list of PI mutations.
- There were no obvious differences in genotype between responders and non-responders among these 20 patients. The prevalence of PI mutations at each position (defined by the LPV mutation score or the DAP) was similar, as was the prevalence of DAP RT mutations.

Table 1. Prevalence of Baseline LPV Mutation Score and DAP PI Mutations (Patients with Baseline PI Mutations at Positions 82, 54, and 10 Plus ≥3 Additional PI Mutations)

| Week 48 | LPV Mutation Score | Number of DAP PI Mutations | Number of DAP NRTI Mutations | NRTI PSS | NRTI GSS | Mean/Median Phenotypic Susceptibility | LPV Mutation Score | | | | | | | | | | | Additional PI Mutations in Modified DAP List | | | | | | | | | DAP NRTI Mutations | | | | | | | | | | | | | | | | | | | |
|---|---|---|---|---|---|---|---|---|---|---|---|---|---|---|---|---|---|---|---|---|---|---|---|---|---|---|---|---|---|---|---|---|---|---|---|---|---|---|---|---|---|---|---|---|---|
| | | | | | | | 10 | 20 | 24 | 46 | 53* | 54 | 63* | 71 | 82 | 84 | 90 | 32 | 33 | 36 | 47 | 48 | 50 | 73 | 77 | 88 | 41 | 62 | 65 | 67 | 69 | 70 | 74 | 75 | 77 | 115 | 116 | 151 | 184 | 210 | 215 | 219 |
| Non-response | 6 | 7 | 2 | 1 | 1 | | I | | | | | V | P | V | A | | M | | | | | V | | | I | | | | | | | | | | | | | | V | | Y | |
| Non-response | 6 | 6 | 3 | 1 | 1 | | I | | | | | V | P | V | T | | M | | | | | | | | I | | L | | | N | | | | | | | | | | | Y | |
| Non-response | 6 | 7 | 6 | 0 | 0 | LPV: | I | | | L | | T | P | L | A | | | | | | | V | | | I | | L | | | N | D | | | | | | | | V | W | Y | |
| Non-response | 6 | 6 | 6 | 0 | 0 | 47.4/50.5 | I | | | I | | V | LP | | F | | M | | | | | | | | I | | L | | | N | D | | | | | | | | V | W | Y | |
| Non-response | 7 | 6 | 5 | 2 | 2 | | FV | | | I | | V | P | V | A | | M | | | | | | | | | | L | | | N | | R | | VIM | | | | | | | F | E* |
| Non-response | 7 | 7 | 5 | 1 | 1 | EFV: | I | | I | I | | V | P | V | A | | | | | | | | | | I | | L | | | N | D | | | | | | | | V | | Y | |
| Non-response | 8 | 7 | 4 | 0 | 0 | 2.8/0.6 | FI | R | I | L | L | V | P | | A | | | | I* | I | | S* | | | | | L | | | | | | LI* | | | | | | MV | W | Y | |
| Non-response | 8 | 7 | 4 | 1 | 2 | | I | R | | | L | V | P | V | A | | M | | | I | | | | | | | L | | | N | | | | M* | | | | | | W | Y | |
| Non-response | 9 | 8 | 5 | 0 | 0 | | I | R | I | LV | FL | V | P | V | A | | | | | I | | | | | | NS* | L | | | N | | R | | | | | | | V | | F | E* |
| Non-response | 9 | 10 | 5 | 0 | 1 | | IV | KR | | | L | V | P | V | VA | IV | M | | F | I | | | | GS | | | | | | N | | G* | | | | | Y | M | | LW | Y | |
| Mean/Median: | 7.2/7 | 7.1/7 | 4.5/5 | 0.6/0.5 | 0.8/1 | Pts (out of 10) with Mutation: | 10 | 4 | 3 | 6 | 4 | 10 | 10 | 8 | 10 | 1 | 6 | 0 | 1 | 4 | 0 | 2 | 0 | 1 | 5 | 0 | 8 | 0 | 0 | 8 | 4 | 2 | 0 | 1 | 0 | 0 | 1 | 1 | 6 | 5 | 10 | 0 |
| Response | 6 | 6 | 3 | 1 | 0 | | LF | | | | | V | P | V | A | | M | | LI* | I | | | | | | | | | | N | | R | | | | | | | V | | | |
| Response | 6 | 5 | 4 | 1 | 0 | | I | | | | L | V | AP | V | A | | | | | | | | | GS | | | | | | N | | G* | | | | | | | V | | F | Q |
| Response | 6 | 6 | 4 | 1 | 0 | LPV: | V | R | | | | V | P | | A | | M | | | I | | | | | | | L | AV | | | | | | | | | | | V | | Y | |
| Response | 7 | 7 | 3 | 2 | 0 | 19.9/16.0 | I | | I | | | V | P | LV | A | V | | | | | | | | | I | | L | | | | | | | | | | | | V | | F | |
| Response | 7 | 6 | 5 | 0 | 0 | | I | | | | | V | P | V | A | V | M | | | | | | | | | | L | | | N | | | | | | | | | MV | W | Y | |
| Response | 7 | 6 | 5 | 0 | 1 | EFV: | I | | I | | FL | V | P | V | T | | | | | I | | | | | | | L | | | | D | | | | | | | | V | W | Y | R* |
| Response | 7 | 9 | 4 | 1 | 2 | 0.3/0.3 | I | | | ML | | V | P | V | A | | M | | F | | | | | ST | I | S* | L | | | N | | | | M* | | | | | | W | Y | |
| Response | 7 | 7 | 6 | 2 | 1 | | I | I* | | L | | V | P | V | A | | M | | LV* | | | | | GS | | | L | | | N | | R | | | | | | | V | | F | Q |
| Response | 8 | 8 | 4 | 0 | 0 | | I | R | | L | | V | P | V | A | | M | | | I | | | | | | | L | | | N | | | | | | | | | V | | Y | |
| Response | 10 | 9 | 4 | 1 | 1 | | I | M | | I | L | V | P | V | VA | IV | M | | | I | | | | | | | L | | | | | | | | | | | | V | W | Y | |
| Mean/Median: | 7.1/7 | 6.9/6.5 | 4.2/4 | 0.9/1 | 0.5/0 | Pts (out of 10) with Mutation: | 10 | 3 | 2 | 4 | 3 | 10 | 10 | 9 | 10 | 3 | 7 | 0 | 1 | 5 | 0 | 0 | 0 | 3 | 2 | 0 | 8 | 1 | 0 | 6 | 1 | 2 | 0 | 0 | 0 | 0 | 0 | 0 | 9 | 4 | 9 | 2 |

* K20I, L33I/V, G48S, F53L, L63P, and N88S not included among DAP PI mutations. K20I not included in LPV mutation score.
K70G, L74I, V75M, and K219E/R not included among DAP NRTI mutations.

- The following factors were analyzed to investigate their association with Week 48 virologic response: phenotypic susceptibility to LPV and EFV, the interaction between the fold IC_{50} of LPV and the fold IC_{50} of EFV, baseline HIV RNA, baseline body weight, number of NRTIs to which the subject was genotypically susceptible, and number of NRTIs to which the subject was phenotypically susceptible (Table 2).
- In these univariate analyses, phenotypic susceptibility to LPV and the product of the LPV and EFV phenotypic susceptibilities were each associated with Week 48 response, suggesting that for complex baseline patterns of multiple mutations, viral phenotype may provide additional predictive power over genotype.

Table 2. Baseline Parameters from Patients with Baseline PI Mutations at Positions 82, 54 and 10 Plus ≥3 Additional Mutations

Baseline Covariate	Responders (n=10)	Non-responders (n=10)	p-value
Median fold IC_{50} of LPV	16	50.5	0.014
Median fold IC_{50} of EFV	0.3	0.6	0.157
Median log baseline HIV RNA	4.14	4.75	0.162
Median baseline body weight	70.5	77.5	0.059
Number of subjects phenotypically susceptible to ≥1 NRTI in regimen	7	5	0.365
Number of subjects genotypically susceptible to ≥1 NRTI in regimen	4	6	0.384
Median fold IC_{50} (LPV) X fold IC_{50} (EFV)	4.6	20.7	0.0028

Mutations Observed Among Other Non-responders

- A total of 12 patients were considered non-responders at Week 48 using a viral load cutoff of 400 copies/mL, including the 10 patients with a LPV mutation score of 6 or more discussed above and 2 patients with LPV mutation scores ≤5. Baseline genotypes for each of these 2 patients are given in Table 3.
- One of these 2 patients displayed a baseline pattern based on the primary L90M mutation (with no mutation at position 82).

Table 3. Prevalence of Baseline LPV Mutation Score and DAP PI Mutations (Non-responders with 5 or Fewer Baseline LPV Mutations)

Week 48	LPV Mutation Score	Number of DAP PI Mutations	LPV Mutation Score											Additional PI Mutations in Modified DAP List								
			10	20	24	46	53*	54	63*	71	82	84	90	32	33	36	47	48	50	73	77	88
Non-response	2	2				MI			P												I	
Non-response	4	5	LI	R				IV	LH				LM			I						

* F53L and L63P not included among DAP PI mutations.

- Five patients who were responders in the analysis at <400 copies/mL at Week 48 had ultrasensitive assay results above 50 copies/mL. Baseline genotypes for these 5 patients are given in Table 4.
- Four of these 5 non-responders had either L90M or I84V at baseline, and only one of these patients also had a V82 mutation.

Table 4. Prevalence of Baseline LPV Mutation Score and DAP PI Mutations (Non-responders Using Ultrasensitive Assay)

Week 48 (HIV RNA >50 copies/mL)	LPV Mutation Score	Number of DAP PI Mutations	LPVMutation Score											Additional PI Mutations in Modified DAP List								
			10	20	24	46	53*	54	63*	71	82	84	90	32	33	36	47	48	50	73	77	88
Non-response	1	1							P							I						
Non-response	4	5	I	I*					P	I			M							S	I	
Non-response	5	6	I			I			P	T			M							A	I	
Non-response	6	6	LF					V	P	V	A		M		LI*	I						
Non-response	7	8	I	I*	I	I		V	P	V		V								S	I	

* K20I, L33I/V, F53L, and L63P not included among DAP PI mutations. K20I not included in LPV mutation score.

DISCUSSION

The virologic response at Week 48 in Study M98-957 was statistically associated with the baseline lopinavir mutation score. These results confirm earlier observations of response at Week 24 (Kempf 2000b). All of the isolates from patients with mutation score ≥6 who experienced virologic failure at Week 48 exhibited a common pattern at positions 82, 54, 10 plus at least 3 additional mutations. However, an equal number of patients with a very similar genotypic pattern experienced virologic response. Between these two sets of patients, the baseline phenotypic susceptibility to lopinavir was a significant predictor of response. These results suggest that for complex genotypic patterns involving multiple protease mutations, viral phenotype may provide additional predictive power over genotype.

This analysis has several limitations.

- All patients in this study were NNRTI-naive at baseline and received EFV, which is likely to have improved the virologic response rate, particularly in patients having viruses with multiple mutations and reduced susceptibility to lopinavir. Although susceptibility to EFV was not statistically associated with virologic response, the product of the fold IC_{50} of LPV and EFV was a better predictor of response than the fold IC_{50} of LPV alone in the above subset of subjects with multiple mutations.
- Most patients in this study (50/57, 88%) also received at least 1 NRTI to which they were considered genotypically or phenotypically (<2.5-fold reduction compared to wt) susceptible.
- The relatively small sample size of this study limits the power of this analysis to detect potentially important relationships with response. For example, the only individual mutations that were statistically associated with virologic failure were at positions 54 and 82. Analyses of larger populations of patients may be required to demonstrate associations of other mutations with virologic failure (Calvez, Abstract #82).

The common mutation pattern (positions 82, 54 and 10) among subjects failing therapy with mutation score ≥6 suggests that these mutations are the most important components of the lopinavir mutation score for predicting virologic failure. However, the presence of the L90M mutation, in absence of a mutation at position 82, in isolates from several patients who either failed therapy with mutation score <6 or displayed HIV RNA <400 copies/mL but >50 copies/mL also suggests a significant contribution of other mutations to a lack of virologic response. The presence of the L90M mutation also appears to compromise the genetic barrier to resistance to LPV/r in patients who do not experience complete suppression of replication (Molla, Poster 064).

REFERENCES

R Stryker, S Brun, M King and others. Kaletra (ABT-378/ritonavir) in Antiretroviral-Naive HIV+ Patients: Follow-up Beyond Two Years and Viral Load Suppression Below 3 Copies/mL. 5th International Congress on Drug Therapy in HIV Infection, Glasgow, UK, October 22-26, 2000.

D Kempf, J Isaacson, M King and others. Genotypic Correlates of Reduced *In Vitro* Susceptibility to ABT-378 in HIV Isolates from Patients Failing Protease Inhibitor Therapy. 4th International Workshop on HIV Drug Resistance and Treatment Strategies, Sitges, Spain, June 2000. Abstract 38.

D Kempf, S Brun, R Rode, and others. Identification of Clinically Relevant Phenotypic and Genotypic Breakpoints for ABT-378/r in Multiple PI-Experienced, NNRTI-Naïve Patients. 4th International Workshop on HIV Drug Resistance and Treatment Strategies, Sitges, Spain, June 2000. Abstract 89.

V Calvez, I Cohen-Codar, A G Marcelin and others. Identification of Individual Mutations in HIV Protease Associated with Virologic Response to Lopinavir/ritonavir Therapy. 5th International Workshop on HIV Drug Resistance and Treatment Strategies, Scotsdale, Arizona USA, June 4-8, 2001. Abstract #82.

A Molla, S Brun, K Garren and others. Patterns of Resistance to Lopinavir in PI-Experienced Patients Following Viral Rebound During Lopinavir/ritonavir Therapy. 5th International Workshop on HIV Drug Resistance and Treatment Strategies, Scotsdale, Arizona USA, June 4-8, 2001. Abstract 064.

E011680C