TUPE0241

Analysis of US costs of full virologic suppression for treatment-experienced, HIV-infected patients in the DUET trials

Silas Martin,[1] Andrew M Hill[2]

[1]Johnson & Johnson Pharmaceutical Services LLC, Mechelen, Belgium; [2]University of Liverpool, Liverpool, UK

Silas Martin, MS
Johnson & Johnson
Pharmaceutical Services LLC
Mechelen
Belgium
smartinc@janbe.jnj.com

Abstract

Background

The aim of antiretroviral (ARV) treatment is long-term suppression of HIV RNA below 50 HIV RNA copies/mL. The DUET-1 and DUET-2 trials evaluated the efficacy of a next-generation NNRTI etravirine (ETR; TMC125) versus placebo, given with a background regimen (BR) of NRTIs, darunavir/ritonavir (DRV/r) and optional enfuvirtide (ENF), in treatment-experienced patients.

Methods

Published US ARV treatment costs (MedSpan Price Check PC) were used. Rates of HIV suppression <50 copies/mL in different treatment groups were analyzed in combination with drug costs to calculate the cost per patient with HIV RNA <50 copies/mL.

Results

For the DUET-1 and DUET-2 trials, the average annual per patient cost of ARVs in the ETR arm was $43,993, with 29% of the total cost from NRTIs, 26% from protease inhibitors (PIs), 27% from ENF and 18% from ETR; the mean overall cost in the placebo arm was $35,905. The cost per patient with HIV RNA <50 copies/mL was $72,120 for the ETR arm (61% response at Week 48) vs $89,762 for the placebo arm (40% response at Week 48). For a fixed treatment budget of $1 million, this would lead to 13.9 patients showing HIV RNA suppression if given ETR + BR, vs 11.1 patients given placebo + BR.

Conclusions

In the DUET trials, treatment with ETR was associated with significant reductions in the cost per patient with HIV RNA <50 copies/mL. There was no significant difference in adverse event (AE) rates between arms, but there was a lower rate of progression to AIDS in the ETR arm, which could also influence value assessments.

Please note that some of the data in the abstract have been updated since submission.

Introduction

- The DUET-1 and DUET-2 trials evaluated the efficacy of the next-generation NNRTI ETR vs placebo, given with a BR of NRTIs, DRV/r and optional ENF, in highly treatment-experienced patients
- HIV RNA suppression <50 copies/mL (undetectability) is the primary aim of ARV treatment in both naïve and treatment-experienced patients[1]
- Considering healthcare cost constraints in the treatment of HIV, it is important to show the value of each component of highly active antiretroviral therapy (HAART)

DUET study design and major inclusion criteria

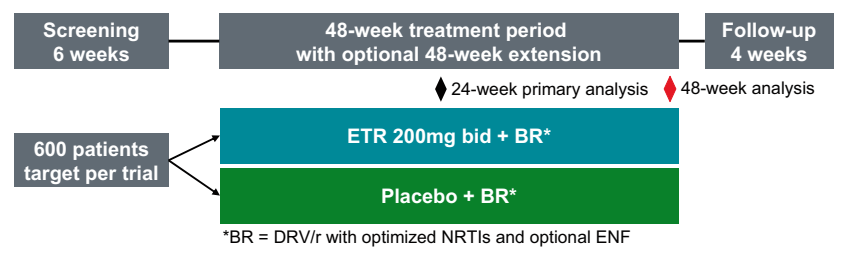

Screening
6 weeks
48-week treatment period
with optional 48-week extension
Follow-up
4 weeks
24-week primary analysis
48-week analysis
600 patients
target per trial
ETR 200mg bid + BR*
Placebo + BR*

*BR = DRV/r with optimized NRTIs and optional ENF

- DUET-1 and DUET-2 differed only in geographic location; pooled analysis was prespecified
- Major inclusion criteria
 - plasma viral load >5000 HIV-1 RNA copies/mL and stable therapy for ≥8 weeks
 - ≥1 NNRTI mutation at screening or in documented historic genotype
 - ≥3 primary PI mutations at screening
- Patients were recruited from Thailand, Australia, Europe and the Americas

Treatment cost calculations in the pooled DUET trials

- Annual US costs of ARVs (Tables 1 and 2) and data on actual ARV usage in the DUET trials (Tables 3 and 4) were used to calculate the total annual cost of treatment for the ETR and placebo groups
- Treatment costs were divided into four categories
 - nucleoside analogs (ZDV, 3TC, ddI, d4T, ABC, TDF, FTC)
 - PIs (DRV/r)
 - fusion inhibitors (ENF)
 - NNRTIs (ETR)
- For this analysis, patients were assumed to continue taking all treatments assigned at baseline for a full 52 weeks

ZDV = zidovudine; 3TC = lamivudine; ddI = didanosine; d4T = stavudine; ABC = abacavir
TDF = tenofovir; FTC = emtricitabine

Annual US costs of NRTIs and ETR

Table 1

Class	Drug name			Dose	Annual cost* (US$)
NRTIs	Epivir®	Lamivudine	3TC	300mg qd	3923
	Emtriva®	Emtricitabine	FTC	200mg qd	3999
	Retrovir®	Zidovudine	ZDV	300mg bid	4584
	Videx® EC	Didanosine	ddI	400mg qd	3988
	Viread®	Tenofovir	TDF	300mg qd	6719
	Zerit®	Stavudine	d4T	40mg bid	4447
	Ziagen®	Abacavir	ABC	300mg bid	5271
NNRTIs	Intelence™	Etravirine	ETR	200mg bid	7957

*Source: Published US antiretroviral treatment costs (MedSpan Price Check PC)
qd = once daily; bid = twice daily

Annual US costs of PIs and ENF

Table 2

Class	Drug name			Dose	Annual cost*: Unboosted PI cost (US$)		Annual cost*: Boosted PI cost (US$)
PIs	Reyataz®	Atazanavir	ATZ	300mg qd	10,011	+ RTV‡	10,635
	Lexiva®	Fosamprenavir	FPV	700mg bid	7448	+ RTV§	8696
	Crixivan®	Indinavir	IDV	800mg bid	3723	+ RTV§	4972
	Invirase®	Saquinavir	SQV	1000mg bid	8827	+ RTV§	10,075
	Kaletra®	Lopinavir	LPV	400mg bid	–	–	8536
	Viracept®	Nelfinavir	NFV	1250mg bid	7659	–	–
	Aptivus®	Tipranavir	TPV	500mg bid	11,410	+ RTV¶	13,906
	Prezista™	Darunavir	DRV	600mg bid	10,002	+ RTV§	11,250
	Norvir®	Ritonavir	RTV	100mg qd	624	–	–
Entry inhibitor	Fuzeon®	Enfuvirtide	T-20/ENF	90mg bid	26,089	–	–

*Source: Published US antiretroviral treatment costs (MedSpan Price Check PC)
‡Ritonavir 100mg qd; §Ritonavir 100mg bid; ¶Ritonavir 200mg bid (public payer price for RTV)

Percentage use of NRTIs and ETR in the ETR and placebo groups of the pooled DUET trials

Table 3

Class	ARV	All patients: ETR + BR (N=599) n	ETR + BR (N=599) %	Placebo + BR (N=604) n	Placebo + BR (N=604) %
NRTIs*	3TC 300mg qd	345	57.8	331	54.8
	FTC 200mg qd	173	29.0	187	31.0
	ZDV 300mg bid	221	37.0	218	36.1
	ddI 400mg qd	117	19.6	129	21.4
	TDF 300mg qd	453	75.9	441	73.0
	d4T 40mg bid	94	15.7	79	13.1
	ABC 300mg bid	127	21.3	130	21.5
NNRTIs	ETR 200mg bid	597	100.0	0	0.0

Data are from the 48-week pooled analysis of DUET
*No statistical differences were observed between treatment groups for NRTI use

Percentage use of PIs and ENF in the ETR and placebo groups of the pooled DUET trials

Table 4

Class	ARV	All patients: ETR + BR (N=599) n	ETR + BR (N=599) %	Placebo + BR (N=604) n	Placebo + BR (N=604) %
PIs	ATZ/r 300/100mg qd	3*	0.5	0	0.0
	FPV/r 700/100mg bid	0	0.0	1*	0.2
	IDV/r 800/100mg bid	0	0.0	0	0.0
	SQV/r 1000/100mg bid	1*	0.2	0	0.0
	LPV/r 400/100mg bid	3*	0.5	0	0.0
	NFV 1250mg bid	0	0.0	0	0.0
	TPV/r 500/200mg bid	0	0.0	0	0.0
	DRV/r 600/100mg bid	595	99.7	601	99.5
	ENF	272	45.6	282	46.7

Data are from the 48-week pooled analysis of DUET; *Protocol violations

Analysis of costs per HIV RNA <50 copies/mL response

- The mean cost of ARV treatment in the DUET trials was combined with the efficacy result of the proportion of patients reaching undetectable viral load from the trials
- The mean cost per undetectable viral load was the overall cost of ARV treatment in each group of the trial, divided by the proportion of patients who reached undetectable viral load
- The incremental cost-efficacy ratio (ICER) is calculated as the incremental difference in cost between groups divided by the incremental difference in proportion reaching undetectable viral load between groups

Pooled DUET patient and disease baseline demographics

Parameter	ETR + BR (n=599)	Placebo + BR (n=604)
Patient demographics		
Male, %	90	89
Caucasian, %	70	70
Disease characteristics		
Viral load, $\log_{10}$ copies/mL, median (range)	4.8 (2.7–6.8)	4.8 (2.2–6.5)
CD4 cells, cells/mm^3, median (range)	99 (1–789)	109 (0–912)
CDC category C, %	58	59
Prior ARV use		
NNRTIs used in screening period, %	12	12
10–15 ARVs, %	66	65
DRV/r, %	4	5
Detectable mutations		
≥2 NNRTI RAMs,* %	69	69
≥3 primary PI RAMs,‡ %	97	97

*From extended NNRTI RAM list (Tambuyzer L, et al. EHDRW 2007. Abstract 67)
‡From Johnson et al. Top HIV Med 2005;13:125–31

DUET study: efficacy

- After 48 weeks of treatment
 - 61% of patients achieved an undetectable viral load (<50 copies/mL) in the ETR group, and 40% in the placebo group (p<0.0001)
 - ETR patients displayed a significant increase in CD4 cell count versus placebo, 98.2 and 72.9 cells/mm^3 (p=0.0006)

Annual mean cost of ARVs in the ETR and placebo groups of the pooled DUET trials

- Table 5 shows that the average annual per patient cost of ARVs in the ETR group was $43,993, with 29% of the total cost from NRTIs, 26% from PIs, 27% from ENF and 18% from ETR

Table 5

Class	ETR + BR (N=599) Mean (US$)	ETR + BR (N=599) Percentage of total cost	Placebo + BR (N=604) Mean (US$)	Placebo + BR (N=604) Percentage of total cost	Difference (ETR-placebo) Mean (US$)
NRTIs	12,824	29.2	12,516	34.9	308
NNRTIs	7957	18.1	0	–	7957
PIs*	11,326	25.7	11,208	31.2	117
ENF	11,886	27.0	12,181	33.9	–294
Total	**43,993**	–	**35,905**	–	**8088**

*The main PI used was DRV/r 600/100mg bid

Annual mean cost of ARVs in the ETR and placebo groups of the pooled DUET trials

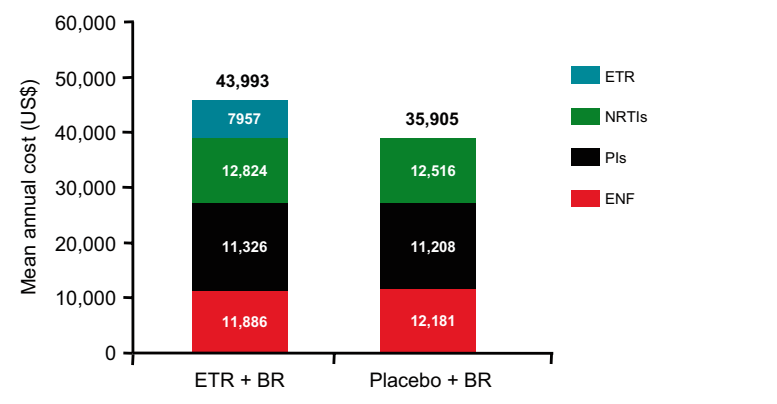

60,000
50,000
40,000
30,000
20,000
10,000
0
Mean annual cost (US$)
43,993
7957
12,824
11,326
11,886
35,905
12,516
11,208
12,181
ETR + BR
Placebo + BR
ETR
NRTIs
PIs
ENF

BR of DRV/r, NRTIs and optional ENF

Cost per efficacy response

- Table 6 shows the cost per efficacy response in the different treatment groups and subgroups including an analysis of
 - the number of patients who could be treated with a fixed $1 million budget for each of the populations analyzed
 - the expected number of patients who could show suppression of HIV RNA <50 copies/mL for this fixed budget

Mean cost per patient with HIV RNA <50 copies/mL at Week 48 in the pooled DUET trials

- Although the total cost of HAART is 23% higher in the ETR group, the higher efficacy in this group leads to a lower cost per person with HIV RNA <50 copies/mL at Week 48

Table 6

All patients	ETR (N=599)	Placebo (N=604)	Difference (ETR-placebo)
Treatment costs (US$)	43,993	35,905	8088
No. treated for $1,000,000	22.7	27.9	–5.1
Undetectable (%)	61	40	21
No. becoming undetectable*	13.9	11.1	2.8
Cost per undetectable (US$)	72,120	89,762	–
ICER (US$)	–	–	38,517

ICER: Incremental Cost Efficacy Ratio = total additional cost / % rise in efficacy

*Number of patients treated for US$1 million who were expected to achieve undetectable viral load based on DUET studies

Conclusions

- Treatment with ETR led to a significantly higher percentage of patients with an undetectable viral load (HIV RNA <50 copies/mL) at Week 48, compared with placebo (61% vs 40%, p<0.0001.[2,3]
- Total cost of ETR-based HAART was $43,993, 23% higher than the cost of placebo-based HAART ($35,905), however the higher efficacy in the ETR group leads to a lower cost per patient with undetectable viral load at Week 48.
- ETR-based HAART was cost-effective versus placebo + BR in this analysis, with respect to
 - the cost per patient with an undetectable viral load (<50 copies/mL) at Week 48
 - the number of patients who could achieve an undetectable viral load for a fixed $1 million budget.

References

1. DHHS Guidelines. January 29, 2008. Available at: http://aidsinfo.nih.gov.
2. Haubrich R, et al. CROI 2008. Poster 790.
3. Johnson M, et al. CROI 2008. Poster 791.

Acknowledgments

- We express our gratitude to the patients who participated in the studies, as well as the study center staff, the Data Safety Monitoring Boards, Tibotec personnel and the following principal investigators:

DUET-1

Argentina: H Ariza, J Benetucci, L Calanni, L Cassetti, J Corral, D David, A Krolewiecki, M Losso, P Patterson, R Teijeiro; **Brazil:** CA da Cunha, E Kallas, E Netto, JH Pilotto, M Schechter, J Suleiman, A Timerman; **Chile:** J Ballesteros, R Northland; **Costa Rica:** A Alvilés Montoya, G Herrera Martinez, A Solano Chinchilla; **France:** M Dupon, C Katlama, JM Livrozet, P Morlat, C Piketty, I Poizot-Martin; **Mexico:** J Andrade-Villanueva, G Reyes-Terán, J Sierra-Madero; **Panama:** A Canton, A Rodriguez, N Sosa; **Puerto Rico:** JO Morales Ramirez, JL Santana Bagur, R Soto-Malave; **Thailand:** T Anekthananon, P Mootsikapun, K Ruxrungtham; **USA:** M Albrecht, N Bellos, R Bolan, P Brachman, C Brinson, F Cruickshank, R Elion, WJ Fessel, T Hawkins, S Hodder, T Jefferson, H Katner, C Kinder, M Kozal, D McDonough, K Mounzer, D Norris, W O'Brien, G Pierone, K Raben, B Rashbaum, M Rawlings, B Rodwick, P Ruane, J Sampson, S Schrader, A Scribner, M Sension, D Sweet, B Wade, D Wheeler, A Wilkin, T Wilkin, T Wills, M Wohlfeiler, K Workowski

DUET-2

Australia: J Chuah, D Cooper, B Eu, J Hoy, C Workman; **Belgium:** N Clumeck, R Colebunders, M Moutschen; **Canada:** J Gill, K Gough, P Junod, D Kilby, J Montaner, A Rachlis, CM Tsoukas, SL Walmsley; **France:** C Arvieux, L Cotte, JF Delfraissy, PM Girard, B Marchou, JM Molina, D Vittecoq, Y Yazdanpanah, P Yeni; **Germany:** S Esser, G Fätkenheuer, H Gellermann, K Göbels, FD Goebel, H Jäger, JK Rockstroh, D Schuster, S Staszewski, A Stoehr; **Italy:** A Antinori, G Carosi, G Di Perri, R Esposito, F Mazzotta, G Pagano, E Raise, S Rusconi, L Sighinolfi, F Suter; **The Netherlands:** PHJ Frissen, JM Prins, BJA Rijnders; **Poland:** A Horban; **Portugal:** F Antunes, M Miranda, J Vera; **Spain:** P Domingo, G Garcia, JM Gatell, J González-Lahoz, J López-Aldeguer, D Podzamczer; **UK:** P Easterbrook, M Fisher, C Orkin, E Wilkins; **USA:** B Barnett, J Baxter, G Beatty, D Berger, C Borkert, C Cohen, M Conant, J Ernst, C Farthing, T File, M Frank, JE Gallant, AE Greenberg, C Hicks, DT Jayaweera, S Kerkar, N Markowitz, C Martorell, C McDonald, D McMahon, M Mogyoros, RA Myers Jr, G Richmond.

Supported by Tibotec

Presented at the XVIIth International AIDS Conference, Mexico City, Mexico, August 3–8 2008.

This poster is available on-line at www.tibotec.com