JD125407 ARTEMIS poster Level 1 30/7/08 15:51 Page 1

THPE0151

ARTEMIS: lipid tolerability and safety of darunavir and lopinavir in antiretroviral-naïve patients

Eduardo Arathoon,[1] Ezio Baraldi,[2] Poh Lian Lim,[3] Carline Vanden Abeele,[4] Magda Opsomer,[4] John Slowick,[5] Frank Tomaka,[5] Ludo Lavreys[4]

[1]Clinica Familiar Luis Angel Garcia, Guatemala City, Guatemala; [2]Trialtech Clinical Research, Pretoria, South Africa; [3]Tan Tock Seng Hospital, Singapore; [4]Tibotec BVBA, Mechelen, Belgium; [5]Tibotec Inc., Yardley, PA, USA

Address correspondence to: Frank Tomaka, MD, Tibotec Inc., 1020 Stony Hill Road, Yardley, PA 19067, USA; Fax: +1 609 730 7513; Email: ftomaka@tibus.jnj.com

Introduction

- The efficacy and tolerability of the protease inhibitor (PI) darunavir (DRV; TMC114) with low-dose ritonavir (DRV/r) 800/100mg qd has been established in treatment-naïve, HIV-1-infected patients in the 48-week primary analysis of the ARTEMIS (TMC114-C211; **A**nti**R**etroviral **T**herapy with TMC114 **E**xa**m**ined **I**n Naïve **S**ubjects) trial[1]
 - 84% of patients receiving DRV/r 800/100mg qd achieved HIV-1 RNA <50 copies/mL at Week 48 vs 78% of patients receiving lopinavir with low-dose ritonavir (LPV/r) 800/200mg total daily dose (p value for noninferiority, p<0.001)
 - patients in the DRV/r arm, compared with those in the LPV/r arm, had a lower incidence of grade 2–4 gastrointestinal adverse events (AEs) at least possibly related to treatment (7% vs 14%, respectively, p<0.01).
- Previous studies have shown that PIs can alter the lipid profiles of HIV-1-infected patients[2,3]
 - although the underlying mechanism is not yet certain, Carr et al. hypothesized that PIs inhibit the activity of two proteins regulating lipid metabolism (cytoplasmic retinoic-acid binding protein type 1 and low-density lipoprotein (LDL) receptor-related protein), which have structural similarities to the target HIV-1 protease.[4]
- A meta-analysis of 12 clinical trials of first-line highly-active antiretroviral therapy suggests that the choice of PI can affect lipid elevations[5]
 - treatment-naïve patients receiving LPV/r or fosamprenavir/ritonavir showed significantly greater rises in triglycerides and cholesterol than DRV/r, saquinavir/ritonavir or atazanavir/ritonavir (p<0.001).
- In this analysis, we compared the lipid-related tolerability of DRV/r 800/100mg qd with that of LPV/r 800/200mg total daily dose in ARTEMIS.

Methods

Study design

- ARTEMIS is an ongoing, randomized, controlled, open-label, Phase III trial designed to compare the efficacy, safety and tolerability of once-daily DRV/r 800/100mg versus LPV/r 800/200mg total daily dose (400/100mg bid or 800/200mg qd depending on local regulatory status and investigator/patient preference) in treatment-naïve adult patients with HIV-1 RNA ≥5000 copies/mL.
- All patients received a fixed background regimen of tenofovir disoproxil fumarate (TDF) 300mg qd and emtricitabine (FTC) 200mg qd.

Assessments and endpoints

- Safety assessments were performed at screening, baseline, Week 2 and every 4 weeks until Week 16, at Week 24 and every 12 weeks thereafter. Patients were required to fast for at least 10 hours prior to blood sampling for biochemistry tests.
- The intent-to-treat (ITT) population was used for the analysis.
- Incidence and severity of AEs and laboratory abnormalities were evaluated throughout the observation period.
- Triglyceride, total cholesterol, LDL (calculated) and high-density lipoprotein (HDL) levels were evaluated.
- Lipid-related parameters were classified as being above or below the US National Cholesterol Education Program (NCEP)[6] cut-offs at any time during the observation period, which was between baseline and a mean of 55 (DRV/r arm) or 53 (LPV/r arm) weeks.
- Patients were allowed to use atorvastatin, rosuvastatin and fibrates.
- Written informed consent was obtained from all patients. Study protocols were reviewed and approved by the appropriate institutional ethics committees and health authorities, and were conducted in accordance with the Declaration of Helsinki.

Results

Patient disposition and baseline characteristics

- A total of 689 patients were randomized to receive once-daily DRV/r 800/100mg (n=343) or LPV/r 800/200mg total daily dose (n=346) plus fixed-dose TDF/FTC qd.
- Baseline characteristics were well balanced between treatment arms (Table 1).

Overall safety

- Most AEs were grade 1 or 2 in severity, and discontinuations due to AEs were infrequent; fewer DRV/r than LPV/r patients discontinued due to an AE (3% vs 7%, post-hoc analysis, p<0.05).

Table 1. Baseline demographics and disease characteristics.

	DRV/r (n=343)	LPV/r (n=346)
Male, n (%)	239 (70)	241 (70)
Mean age, years	35	35
Race, n (%)		
Black	80 (23)	71 (21)
Caucasian/White	137 (40)	153 (44)
Hispanic	77 (22)	77 (22)
Oriental/Asian	44 (13)	38 (11)
Other	4 (1)	5 (1)
Missing	1 (1)	2 (1)
Mean body mass index, kg/m^2	24	24
Disease characteristics		
Mean known duration of infection, years (SD)	2.4 (3.6)	2.5 (3.6)
Mean HIV RNA, $\log_{10}$ copies/mL (SD)	4.86 (0.64)	4.84 (0.60)
Median CD4 cell count, cells/mm^3 (range)	228 (4–750)	218 (2–714)
Hepatitis B and/or C coinfection, n (%)	43 (13)	48 (14)
CDC class, n (%)		
A	226 (66)	217 (63)
B	91 (27)	95 (27)
C	26 (8)	34 (10)
Mean lipid levels, mg/dL (mmol/L)		
Triglycerides	130.72 (1.47)	123.31 (1.39)
Total cholesterol	156.15 (4.00)	156.72 (4.02)
LDL*	91.84 (2.35)	92.63 (2.38)
HDL	39.62 (1.02)	39.80 (1.02)

*Calculated; SD = standard deviation; CDC = Centers for Disease Control and Prevention

Change in mean lipid levels up to Week 48

- For all lipid-related parameters in both treatment groups, small increases were seen at Week 48 compared to baseline (Figures 1a–e).
- Mean triglyceride levels in the DRV/r group remained within the recommended limits of the NCEP criteria throughout the trial, but in the LPV/r group levels were above the limits of the normal range as early as Week 2 of the trial and remained elevated throughout (Figure 1a).
- Despite higher mean levels of total cholesterol over time in the LPV/r group versus the DRV/r group, the mean levels remained within the recommended limits of the NCEP criteria (Figure 1b).
- Changes in LDL, HDL and the total cholesterol/HDL ratio were similar for both treatment groups (Figures 1c–e).

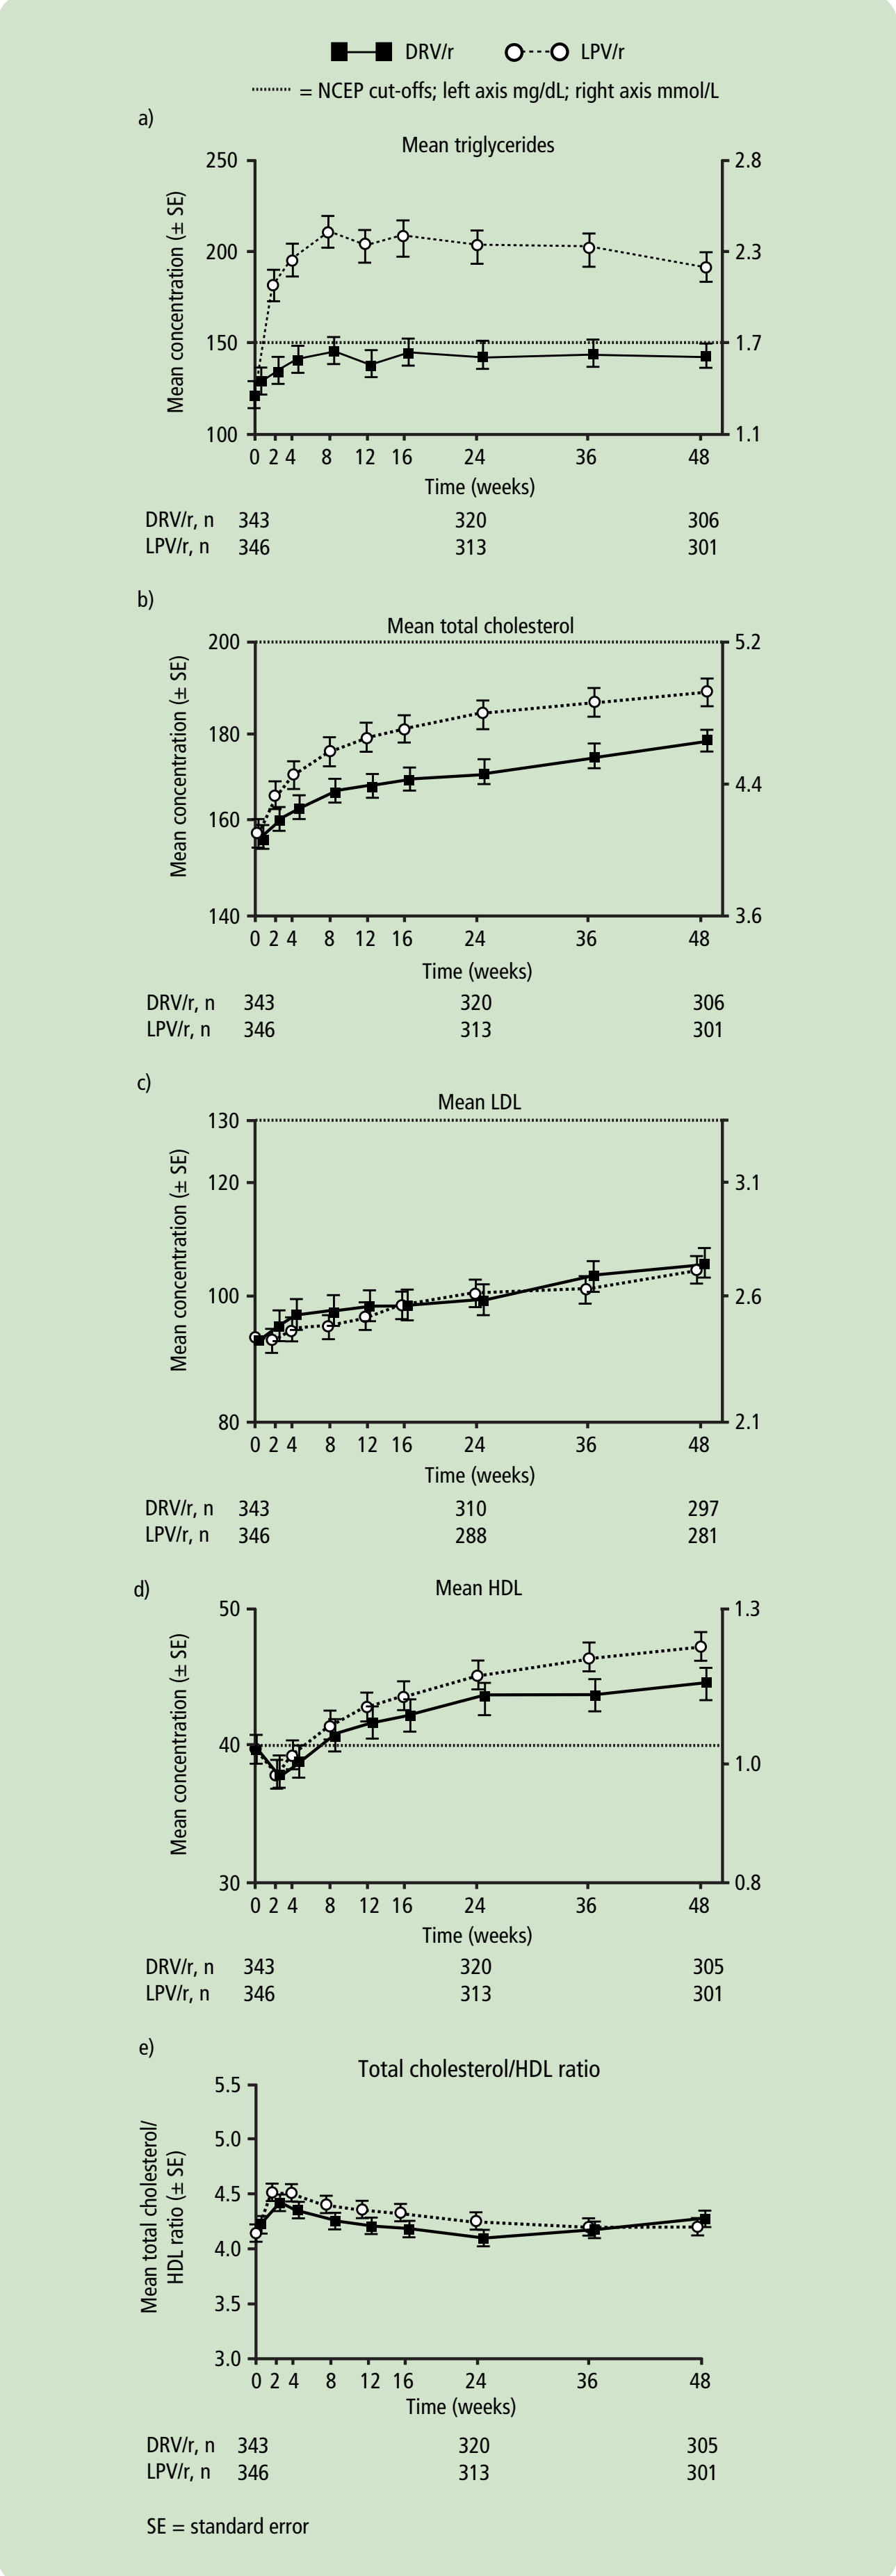
DRV/r
LPV/r
= NCEP cut-offs; left axis mg/dL; right axis mmol/L
a)
Mean triglycerides
Mean concentration (± SE)
Time (weeks)
DRV/r, n 343 320 306
LPV/r, n 346 313 301
b)
Mean total cholesterol
DRV/r, n 343 320 306
LPV/r, n 346 313 301
c)
Mean LDL
DRV/r, n 343 310 297
LPV/r, n 346 288 281
d)
Mean HDL
DRV/r, n 343 320 305
LPV/r, n 346 313 301
e)
Total cholesterol/HDL ratio
Mean total cholesterol/HDL ratio (± SE)
DRV/r, n 343 320 305
LPV/r, n 346 313 301
SE = standard error

Figure 1. Change in mean levels up to Week 48 for a) triglycerides, b) total cholesterol, c) LDL, d) HDL and e) total cholesterol/HDL ratio.

Lipid-related AEs

- Lipid-related AEs were reported in fewer patients in the DRV/r than the LPV/r arm (6% vs 12%; Table 2)
 - this difference was not accounted for by the use of lipid-modifying agents, which were used by similar proportions of patients in the DRV/r (n=21; 6%) and the LPV/r arm (n=26; 8%) during the treatment period.
- The incidence of individual lipid-related AEs was generally lower in the DRV/r than in the LPV/r group (Table 2).
- No grade 3 or 4 lipid-related AEs were reported as serious AEs.
- No patients in either treatment group permanently discontinued or temporarily interrupted treatment due to lipid-related AEs.

Table 2. Lipid-related AEs overall and grade 2–4 lipid-related AEs considered at least possibly related to treatment.

	DRV/r (n=343)		LPV/r (n=346)	
Mean exposure, weeks	55		53	
Dictionary-derived term, n (%)*	**Overall regardless of cause**	**Grade 2–4 at least possibly related to treatment**	**Overall regardless of cause**	**Grade 2–4 at least possibly related to treatment**
Any lipid-related AE	*19 (6)*	*12 (3)*	*43 (12)*	*27 (8)*
Hypertriglyceridemia	4 (1)	4 (1)	17 (5)	11 (3)
Hypercholesterolemia	5 (1)	2 (1)	9 (3)	7 (2)
Hyperlipidemia	4 (1)	2 (1)	9 (3)	5 (1)
Blood cholesterol increased	2 (1)	1 (0)	5 (1)	2 (1)
LDL increased	4 (1)	4 (1)	3 (1)	2 (1)
Blood triglycerides increased	2 (1)	0	1 (0)	1 (0)
Dyslipidemia	1 (0)	1 (0)	2 (1)	1 (0)
Apolipoprotein A-I	0	0	1 (0)	0

*Terms for lipid-related AEs were listed verbatim from investigator reports and therefore values for similar terms could not be combined

Lipid-related laboratory abnormalities

- Overall, lipid-related laboratory abnormalities were observed in both treatment groups.
- The majority of lipid-related laboratory abnormalities were grade 1 or 2 in severity.
- Increased total cholesterol was the most frequent lipid-related laboratory abnormality (33% in the DRV/r group and 45% in the LPV/r group).
- Grade 2–4 increases in triglycerides and total cholesterol were observed less frequently in the DRV/r group (3% and 13%, respectively) than in the LPV/r group (11% and 23%, respectively; Table 3). The proportion of patients with increases in LDL and decreases in HDL was similar between the treatment groups.

Table 3. Treatment-emergent lipid-related laboratory abnormalities.

Laboratory parameter, n (%)*			DRV/r (n=343)	LPV/r (n=346)
Mean exposure, weeks			55	53
Graded	**Worst grade‡**	**Defining level, mg/dL (mmol/L)**		
Triglycerides	2	500–750 (5.65–8.48)	5 (1)	21 (6)
	3	751–1200 (8.49–13.56)	4 (1)	14 (4)
	4	>1200 (>13.56)	1 (0)	3 (1)
Total cholesterol	1	200–239 (5.18–6.19)	69 (20)	77 (22)
	2	240–300 (6.20–7.77)	40 (12)	64 (19)
	3	>300 (>7.77)	4 (1)	14 (4)
LDL	1	130–159 (3.37–4.12)	59 (17)	67 (20)
	2	160–190 (4.13–4.90)	39 (11)	21 (6)
	3	≥190 (≥4.91)	5 (1)	15 (4)
Non-graded				
HDL	Below normal	<40 (<1.03)	57 (17)	65 (19)

*The number of patients with data can vary per parameter, but the percentage reflects the true percentage of observed abnormalities; ‡Based on the Division of AIDS (DAIDS) Table for Grading the Severity of Adult and Pediatric Adverse Events 2004, which does not have a grade 1 classification for triglycerides and grade 4 for total cholesterol and LDL

- Treatment-emergent abnormalities in total cholesterol and triglycerides classified according to NCEP criteria were fewer in the DRV/r than in the LPV/r group at Week 48 (Table 4).
- There was no difference between the treatment groups with respect to the incidence of abnormally high LDL or abnormally low HDL levels.
- Median total cholesterol/HDL ratios at Week 48 remained similar to baseline in both groups (Table 4).

Table 4. Treatment-emergent lipid-related laboratory abnormalities of interest at Week 48 according to NCEP criteria.

Laboratory parameter, n (%)	NCEP criteria mg/dL (mmol/L)	DRV/r (n=343)	LPV/r (n=346)
Triglycerides	High: ≥150 (≥1.69)	126 (37)	183 (54)
Total cholesterol	High: ≥200 (≥5.13)	97 (28)	131 (38)
LDL	High: ≥130 (≥3.33)	85 (25)	88 (26)
HDL	Low: male: ≤40 (1.03); female: ≤50 (1.28)	64 (19)	66 (19)
Median total cholesterol/HDL ratio			
Baseline		4.02	3.94
Week 48		4.09	3.97

Conclusions

- Treatment with once-daily DRV/r 800/100mg was effective and associated with smaller increases in triglycerides and total cholesterol than treatment with LPV/r.
- Patients treated with once-daily DRV/r 800/100mg developed fewer lipid-related AEs compared with those patients who received LPV/r.
- Treatment differences were not accounted for by use of lipid-modifying agents, which were used by similar proportions of patients.
- Once-daily DRV/r 800/100mg is well tolerated and has a more favorable lipid profile than LPV/r in treatment-naïve, HIV-infected patients.

References

1. Ortiz R, et al. AIDS 2008;22:1389–97.
2. Tsiodras S, et al. Arch Intern Med 2000;160:2050–6.
3. Penzak SR, et al. Scand J Infect Dis 2000;32:111–23.
4. Carr A, et al. Lancet 1998;351:1881–3.
5. Hill A, et al. 14th Annual Conference of the British HIV Association (BHIVA), Belfast, UK, 23–25 April 2008. Abstract P79.
6. National Cholesterol Education Program (NCEP) Expert Panel on Detection, Evaluation, and Treatment of High Blood Cholesterol in Adults (Adult Treatment Panel III). Circulation 2002;106:3143–421.
7. AIDS Clinical Trials Group. Division of AIDS table for grading the severity of adult and pediatric adverse events [Online]. 28 December 2004 [accessed 13 June 2008]. Available from: http://www3.niaid.nih.gov/research/resources/DAIDSClinRsrch/PDF/Safety/DAIDSAEGradingTable.pdf.

Supported by Tibotec

Presented at the XVIIth International AIDS Conference, Mexico City, Mexico, August 3–8 2008.

This poster is available on-line at www.tibotec.com.