121

Determination of phenotypic clinical cut-offs for etravirine (ETR): pooled Week 24 results of the DUET-1 and DUET-2 trials

Monika Peeters,[1] Steven Nijs,[1] Johan Vingerhoets,[1] Lotke Tambuyzer,[1] Brian Woodfall,[1] Marie-Pierre de Bethune,[1] Gaston Picchio[2]

[1]Tibotec BVBA, Mechelen, Belgium; [2]Tibotec Inc., Yardley, USA

Monika Peeters
Tibotec BVBA
Generaal De Wittelaan
L11 B3
B2800, Mechelen
Belgium
mpeeter7@tibbe.jnj.com

Abstract

Background

DUET-1 and DUET-2 are ongoing, randomised, placebo-controlled, double-blind, Phase III trials, demonstrating superior antiretroviral activity at 24 weeks of the NNRTI etravirine (ETR; TMC125) + background regimen (BR; darunavir with low-dose ritonavir [DRV/r] + NRTIs ± enfuvirtide [ENF]) versus placebo + BR in treatment-experienced patients. Phenotypic clinical cut-offs (CCOs) for ETR are presented.

Methods

In pooled DUET, 599 patients received ETR. Phenotypic CCOs for antivirogram were determined using analysis of covariance (ANCOVA) models and data-mining techniques in patients not using for the first-time (*de novo*) ENF and excluding those who discontinued before 24 weeks for reasons other than virologic failure (n=403).

Results

Baseline ETR fold-change in 50% effective concentration (FC) was a significant predictor of response (HIV-1 RNA <50 copies/mL) at 24 weeks. Baseline FC and responses to ETR were characterised by a continuum rather than a bimodal distribution. Inverse prediction of the ANCOVA model, with covariates baseline viral load (VL), baseline CD4 cell count and baseline DRV FC, NRTI sensitivity and ETR FC, resulted in an initial CCO of 13, based on a 1 log greater response at Week 24 versus placebo. Since response in patients with baseline FC >13 was still substantial (37%), this value was considered an intermediate CCO. An FC value above which ETR provided no or little additional efficacy benefit (high CCO) could not reliably be established. Data-mining techniques allowed determination of a lower CCO of 3, below which patients exhibited the highest response rate. At baseline, 67%, 18% and 15% of patients had ETR FC ≤3, 3–13, and >13, respectively. At Week 24, 71%, 50% and 37% of patients with FC ≤3, 3–13, and >13, respectively, reached VL <50 copies/mL.

Conclusions

Response in the ETR arms of the DUET trials decreased with increasing baseline ETR FC. The highest response rate was observed in the group of patients with ETR FC ≤3 (lower CCO). The robust responses observed in a substantial number of patients with baseline ETR FC >13 (intermediate CCO) and the low number of observations in this subgroup did not allow for the determination of a high CCO. These CCOs provide phenotypic guidance for use of ETR in treatment-experienced HIV-1-infected patients.

Please note some of the data in the abstract have been updated in the poster

DUET-1 and DUET-2: trial design and inclusion criteria

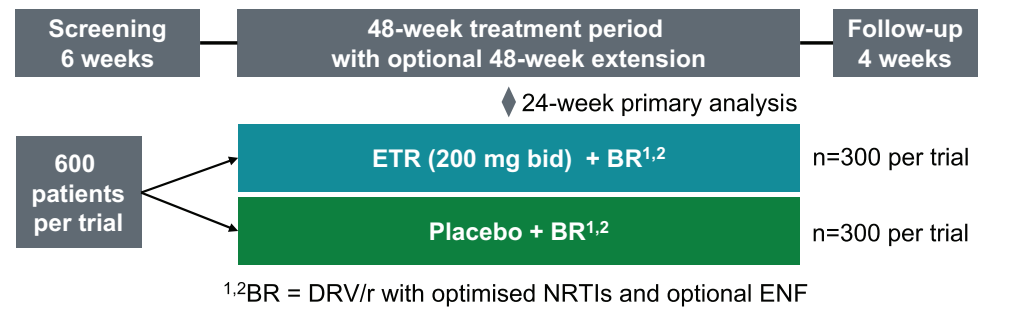
Screening
6 weeks
48-week treatment period
with optional 48-week extension
Follow-up
4 weeks
24-week primary analysis
600
patients
per trial
ETR (200 mg bid) + BR[1,2]
n=300 per trial
Placebo + BR[1,2]
n=300 per trial

[1,2]BR = DRV/r with optimised NRTIs and optional ENF

- Plasma VL at screening: >5,000 HIV-1 RNA copies/mL
- Stable antiretroviral therapy for at least 8 weeks prior to screening and until baseline
- At least three primary protease inhibitor mutations at screening
- At least one NNRTI RAM (either at screening or in documented historical genotype)

RAM = resistance-associated mutation
[1]Madruga JV, et al. Lancet 2007;370:29–38; [2]Lazarin A, et al. Lancet 2007;370:39–48

Patients with VL <50 copies/mL over Week 48 (ITT-TLOVR)

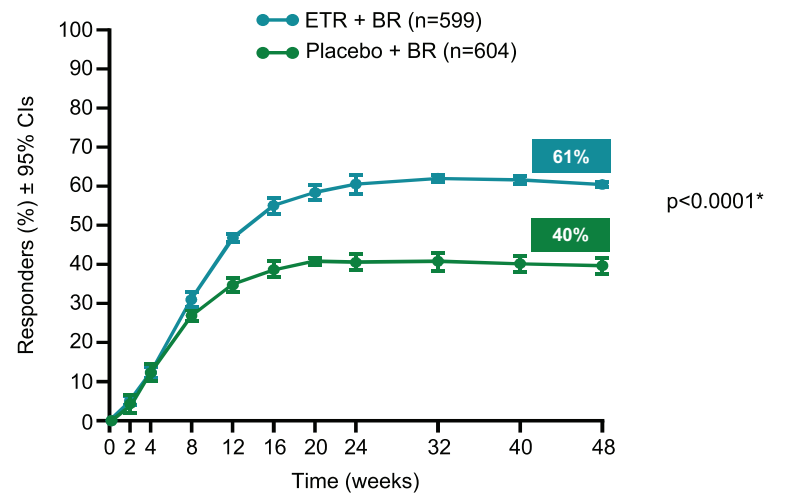
ETR + BR (n=599)
Placebo + BR (n=604)
Responders (%) ± 95% CIs
61%
40%
p<0.0001*
Time (weeks)

- 61% of patients in the ETR group achieved confirmed undetectable VL (<50 copies/mL) compared with 40% in the placebo group at Week 48

ITT = intent-to-treat; TLOVR = time to loss of virologic response; CI = confidence interval; *Logistic regression model

Determination of ETR FC CCO: data used in the analysis

- Pooled DUET-1 and DUET-2 studies – Week 24 response data
- Patients who discontinued for reasons other than virologic failure (non-virologic failure excluded population) were excluded
- Subgroups of patients
 - total PSS* = 0 (n=88): ideal but small sample
 - *de-novo* ENF (n=143)/not *de-novo* ENF (n=403)

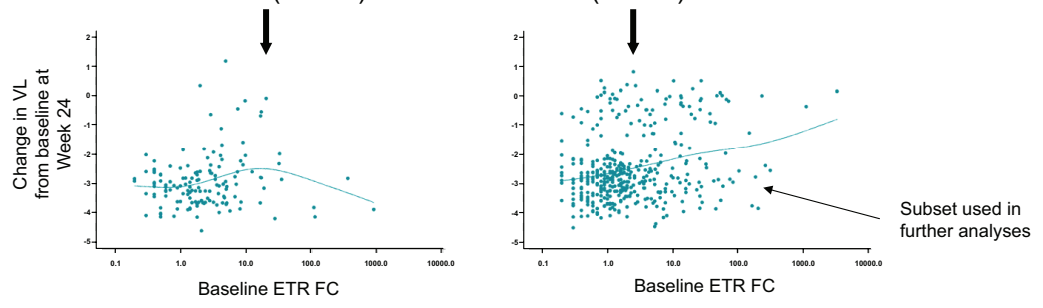
Change in VL from baseline at Week 24
Baseline ETR FC
Subset used in further analyses

LOESS smoothed spline fitted to the raw data

*Phenotypic sensitivity score: ENF counted as 0 if not used *de novo*; DRV counted as 0 if FC >10

Analysis outline: methods

- Phenotypic susceptibility determined by antivirogram
- Efficacy parameters at Week 24
 - VL <50 copies/mL (TLOVR)
 - change in $\log_{10}$ VL (NC=F)
- ANCOVA model for change in $\log_{10}$ VL
 - correction for factors
 - baseline disease characteristics (baseline VL, CD4)
 - baseline resistance (sensitivity of BR: $\log_{10}$ FC ETR, $\log_{10}$ FC DRV, ENF use, number of sensitive NRTIs)
- Data mining using graphical presentations

NC=F = non-completer=failure

Increasing baseline ETR FC was associated with a gradual loss in virologic response at Week 24

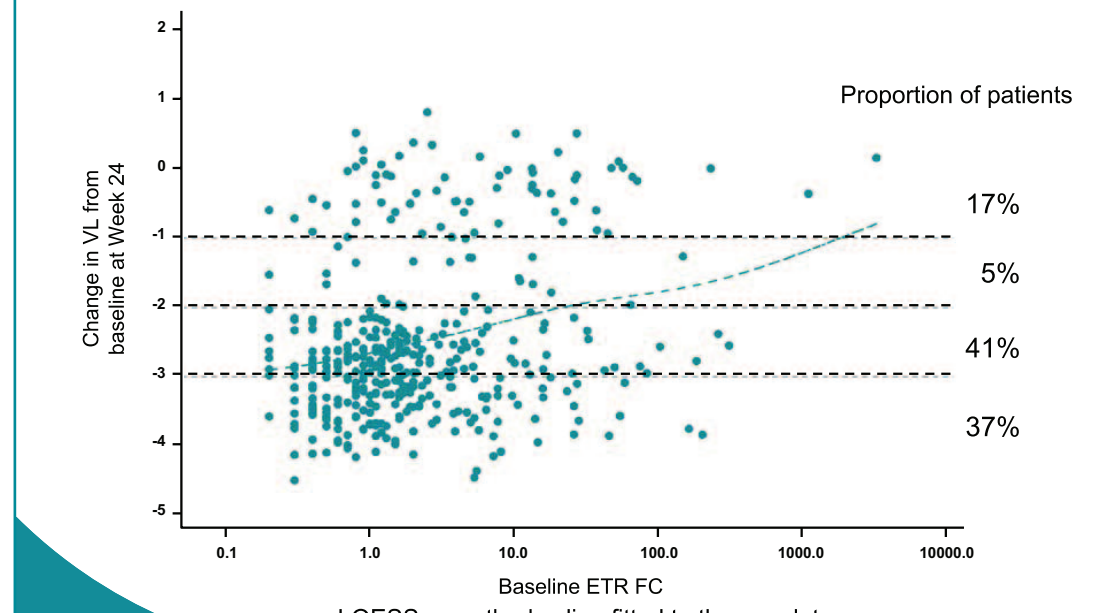
Proportion of patients
17%
5%
41%
37%
Change in VL from baseline at Week 24
Baseline ETR FC

LOESS smoothed spline fitted to the raw data

Change in $\log_{10}$ VL at Week 24 vs ETR FC: confounding factors – high proportion of patients with undetectable VL underestimates the change in VL from baseline

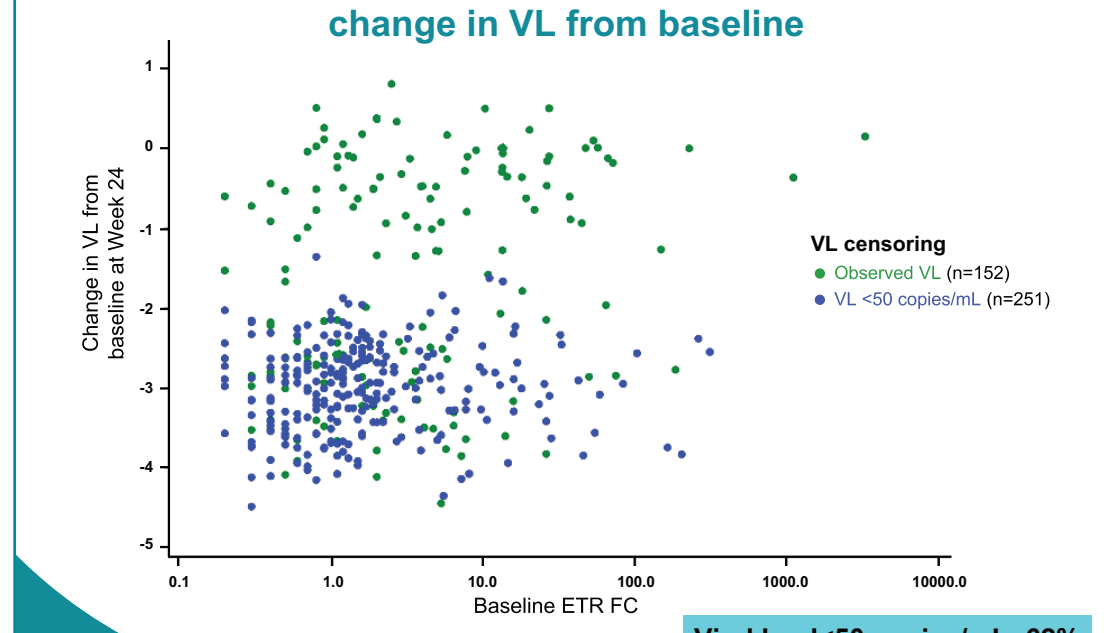
VL censoring
Observed VL (n=152)
VL <50 copies/mL (n=251)
Change in VL from baseline at Week 24
Baseline ETR FC

Viral load<50 copies/mL: 62%

Change in $\log_{10}$ VL at Week 24 vs ETR FC: confounding factors significantly influencing response (cont'd)

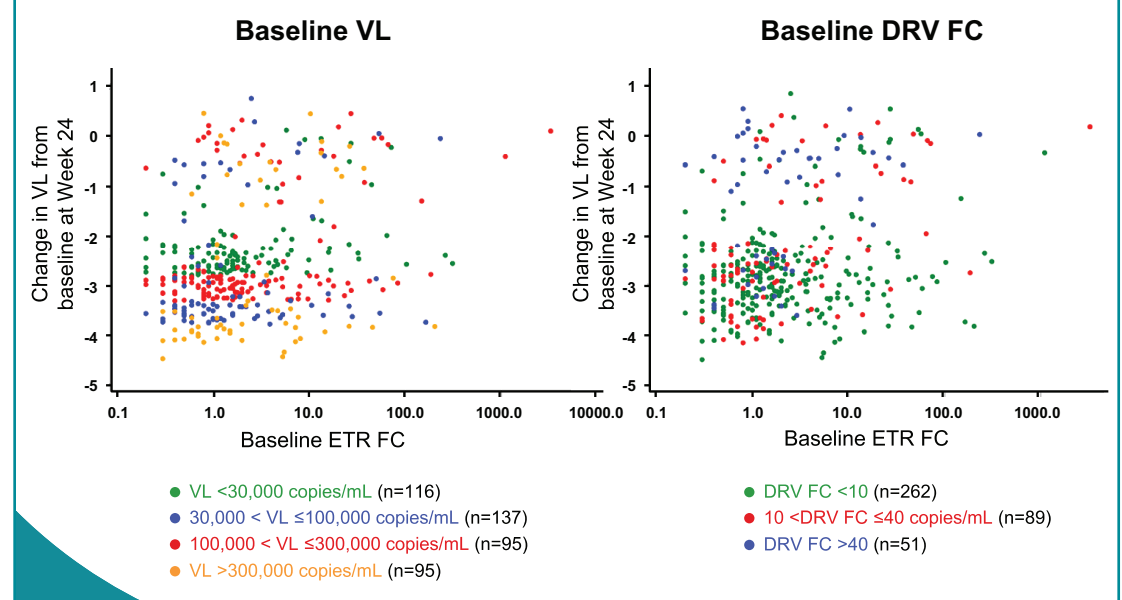
Baseline VL
Baseline DRV FC
Change in VL from baseline at Week 24
Baseline ETR FC
VL <30,000 copies/mL (n=116)
30,000 < VL ≤100,000 copies/mL (n=137)
100,000 < VL ≤300,000 copies/mL (n=95)
VL >300,000 copies/mL (n=95)
DRV FC <10 (n=262)
10 <DRV FC ≤40 copies/mL (n=89)
DRV FC >40 (n=51)

Definition of ETR CCO

- Reference: At Week 24, patients in the placebo arm who were not using ENF *de novo*, had a mean change in $\log_{10}$ VL of –1.4 log
- The ETR CCO was determined as a 1 log better response than placebo = –2.4 log
 - using the ANCOVA model, backward prediction was used to define a cut-off that was associated with at least –2.4 reduction in $\log_{10}$ VL

Change in $\log_{10}$ VL at Week 24 vs ETR FC: identification of CCO at FC=13

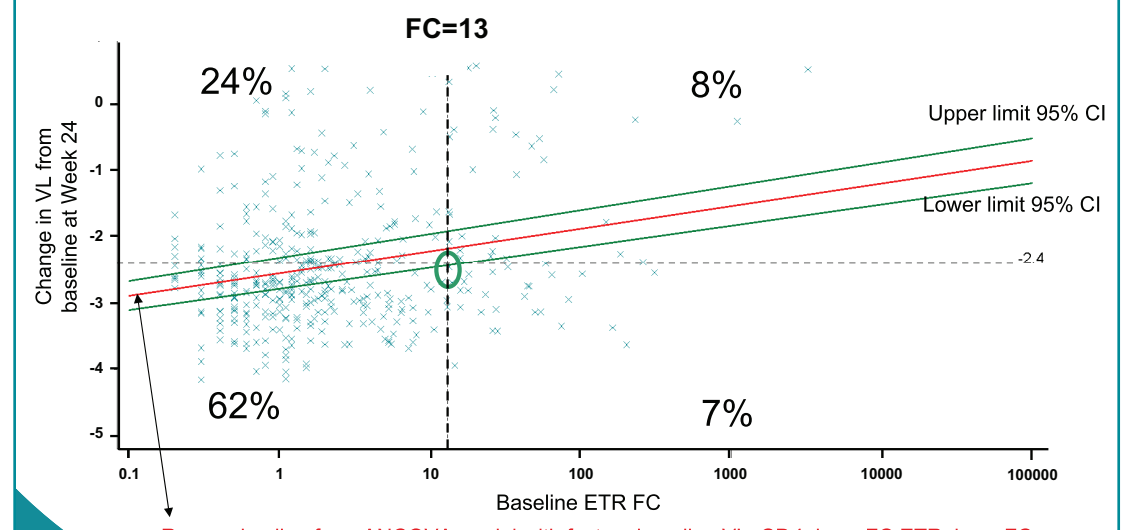
FC=13
24%
8%
62%
7%
Upper limit 95% CI
Lower limit 95% CI
-2.4
Change in VL from baseline at Week 24
Baseline ETR FC

Regression line from ANCOVA model with factors baseline VL, CD4, $\log_{10}$ FC ETR, $\log_{10}$ FC DRV, ENF use, # sensitive NRTIs in ART

Identification of CCOs lower than 13

- Look for baseline ETR FC below which virologic response (<50 copies/mL) is maximal

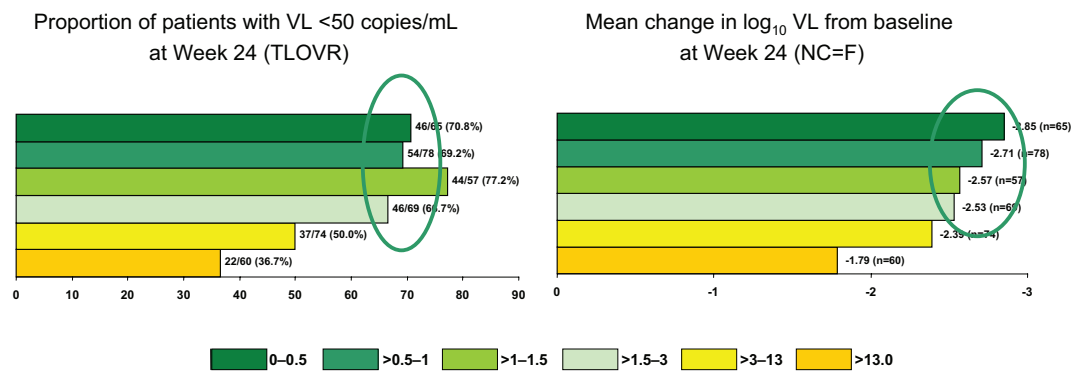
Proportion of patients with VL <50 copies/mL at Week 24 (TLOVR)
Mean change in log10 VL from baseline at Week 24 (NC=F)
0–0.5
>0.5–1
>1–1.5
>1.5–3
>3–13
>13.0

→ Lower CCO = 3

Identification of CCOs higher than 13

- Look for baseline ETR FC above which response (<50 copies/mL) is minimal

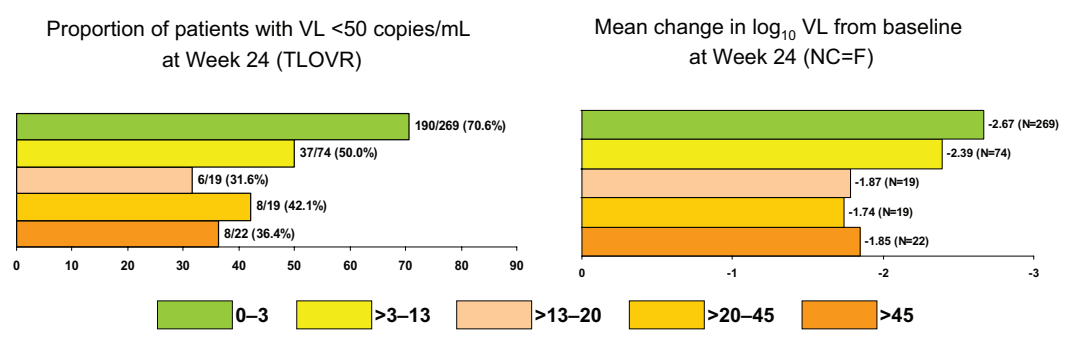
Proportion of patients with VL <50 copies/mL at Week 24 (TLOVR)
Mean change in log10 VL from baseline at Week 24 (NC=F)
190/269 (70.6%)
37/74 (50.0%)
6/19 (31.6%)
8/19 (42.1%)
8/22 (36.4%)
-2.67 (N=269)
-2.39 (N=74)
-1.87 (N=19)
-1.74 (N=19)
-1.85 (N=22)
0–3
>3–13
>13–20
>20–45
>45

- Substantial response was observed above baseline FC=13, thus this represents an intermediate CCO
- Limited data above baseline FC=13, thus a higher CCO could not be established

Confirmation of CCOs 3 and 13

- Optimal cut-off should give best distinction in response between patients with FC <CCO and those with FC >CCO

Difference in proportion of response in patients with FC ≤cut-off and those with FC >cut-off

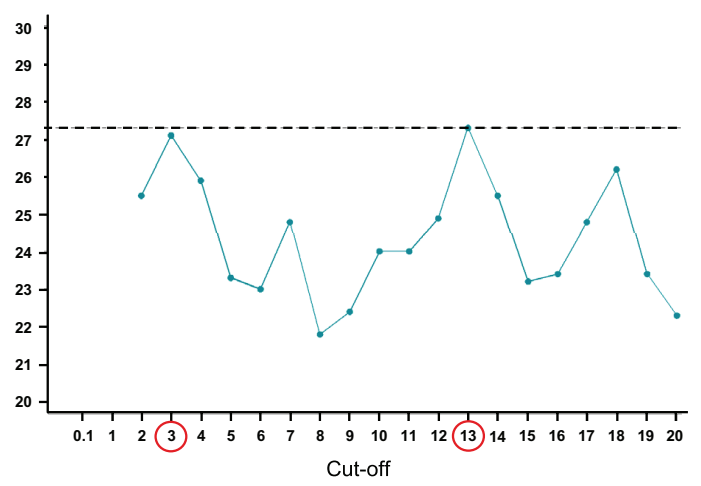
Cut-off

Response according to phenotypic ETR CCOs: pooled Week 24 DUET – patients not using ENF *de novo*

ETR CCO	Proportion of patients with VL <50 copies/mL (TLOVR), % (n)	Mean (SE) decrease in $\log_{10}$ VL from baseline (NC=F)
≤3	71 (190/269)	–2.67 (1.03)
3–13	50 (37/74)	–2.39 (1.21)
>13	37 (22/60)	–1.79 (1.42)
Overall placebo	36 (149/414)	–1.51 (1.42)

Non-virologic failure excluded population

ETR FC vs ETR weighted mutation score – pooled DUET

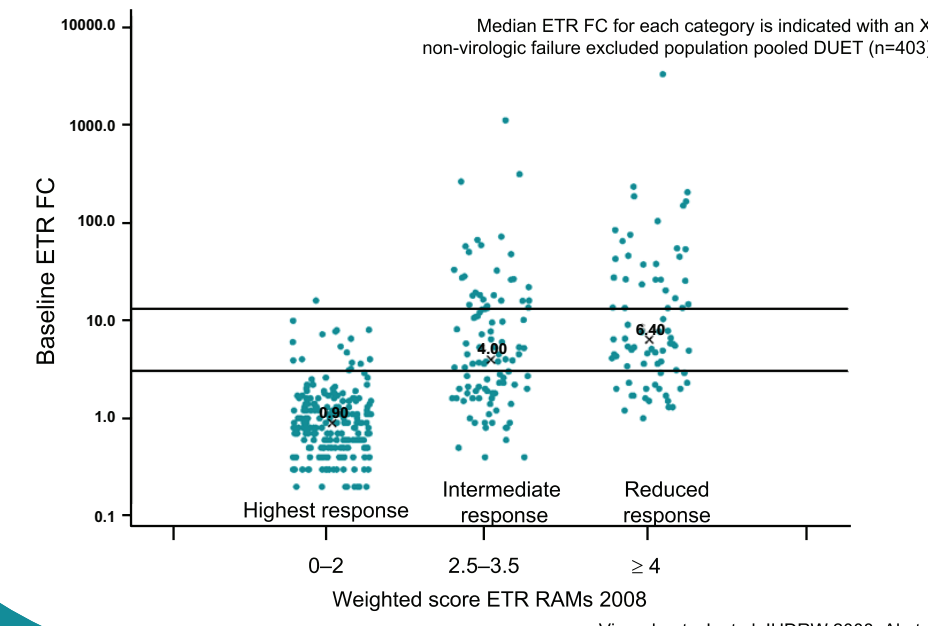
Median ETR FC for each category is indicated with an X
non-virologic failure excluded population pooled DUET (n=403)
Baseline ETR FC
0.90
4.00
6.40
Highest response
Intermediate response
Reduced response
0–2
2.5–3.5
≥ 4
Weighted score ETR RAMs 2008

Vingerhoets J, et al. IHDRW 2008. Abstract 24

Conclusions

- Based on the analysis of the Week 24 DUET virologic response data, CCOs were determined for ETR
- First time that phenotypic CCOs could be determined for an NNRTI
- A lower CCO of 3 and an intermediate CCO of 13 were identified for ETR
 - the 'highest' response rate (71% VL <50 copies/mL) was observed in patients with baseline ETR FC ≤3
 - an 'intermediate' response rate (50% VL <50 copies/mL) was observed in patients with baseline ETR FC between 3 and 13
 - an upper CCO above which patients would no longer benefit from ETR could not yet be determined in this dataset, due to the small number of patients with FC >13 and the substantial virologic response rate in this subset of patients (37% VL <50 copies/mL)
- The majority of patients in DUET had an ETR baseline FC ≤3: 66% (779/1190)

Acknowledgements

- The authors would like to express their gratitude to the following
 - the patients and their families
 - the investigators
 - the study centre staff
 - Virco
 - Tibotec clinical trial staff
 - Tibotec clinical virology, Biometrics

Supported by Tibotec

Presented at the XVIIth International Drug Resistance Workshop, Sitges, Spain, 10–14 June 2008.

This poster is available on-line at www.tibotec.com