Poster No. 1351

Pharmacokinetic interaction between TMC125 and clarithromycin

M Schöller-Gyüre, C Debroye, B Woodfall, F Aharchi, M Peeters, K Vandermeulen, R Hoetelmans

Tibotec BVBA, Mechelen, Belgium

Monika Schöller-Gyüre
Tibotec BVBA
Generaal de Wittelaan L11 B3
B2800, Mechelen
Belgium
mscholle@tibbe.jnj.com

Abstract

Background

TMC125 is an NNRTI with potent activity against both wild-type HIV and viruses resistant to currently approved NNRTIs. TMC125 is a substrate and inducer of CYP3A4. Clarithromycin inhibits CYP3A4 and is converted by CYP3A4 to its active metabolite, 14-OH clarithromycin. This study aimed to evaluate the effect of concomitant use of TMC125 and clarithromycin on the pharmacokinetic (PK) profiles of both agents.

Methods

TMC125-C171 was an open-label, randomized, two-period, crossover trial in HIV-negative volunteers. In Treatment A, 200mg TMC125 bid (Phase III formulation) was given for 8 days. Treatment B started after a washout period of 14 days with the administration of clarithromycin 500mg bid during Days 1–13. TMC125 200mg bid was co-administered in Days 6–13. PK parameters of TMC125 were assessed on Day 8 of Treatment A and Day 13 of Treatment B. PK parameters of clarithromycin and 14-OH clarithromycin were assessed on Days 5 and 13 of Treatment B. PK parameters were analyzed using a linear mixed effect model for crossover design. Safety and tolerability were assessed during the trial.

Results

Sixteen male volunteers participated (median age 29 years). When combined with clarithromycin, TMC125 AUC_{12h} was 142% (90% CI: 134–150%) compared with administration of TMC125 alone. TMC125 C_{max} and C_{min} were 146% (90% CI: 138–156%) and 146% (90% CI: 136–158%), respectively. AUC_{12h}, C_{max} and C_{min} of clarithromycin were 61% (90% CI: 53–69%), 66% (90% CI: 57–77%) and 47% (90% CI: 38–57%), respectively, when combined with TMC125 compared with administration of clarithromycin alone. The AUC_{12h}, C_{max} and C_{min} of 14-OH clarithromycin were 121% (90% CI: 105–139%), 133% (90%CI: 113–156%) and 105% (90% CI: 90–122%), respectively, when given in combination with TMC125. The short-term co-administration of TMC125 and clarithromycin was generally safe and well tolerated.

Conclusions

TMC125 induces the metabolism of clarithromycin, whereas clarithromycin shows an inhibitory effect on TMC125 metabolism. No dose adjustment of TMC125 is recommended. Since 14-OH clarithromycin has reduced activity against *Mycobacterium avium* complex (MAC), overall activity against this pathogen may be altered. Alternatives to clarithromycin such as azithromycin should be considered.

Introduction

- TMC125 is a next generation NNRTI with potent in-vitro activity against both wild-type HIV-1 and HIV-1 resistant to current NNRTIs[1]
- A Phase IIb trial (TMC125-C223) in treatment-experienced HIV patients demonstrated that TMC125, with an optimized background regimen, reduced viral load at 48 weeks significantly more than active control. No dose-related effects on safety and tolerability were noted[2,3]
- TMC125 is predominantly metabolized by CYP3A4, CYP2C and glucuronidation; it is an inducer of CYP3A4 and an inhibitor of CYP2C
- Clarithromycin is a macrolide antibiotic indicated for the treatment of infections caused by susceptible strains of micro-organisms
- Clarithromycin is converted by CYP3A4 to its active metabolite 14-OH clarithromycin; it is an inhibitor of CYP3A enzymes[4]
- To investigate concomitant administration, an interaction study with clarithromycin and TMC125 (Phase III formulation) was conducted

Study design

- TMC125-C171 was a Phase I, open-label, two-way, two-period, crossover trial in 16 HIV-negative volunteers
- Two treatment sessions (A and B) were scheduled for all volunteers, separated by a washout period of at least 14 days. Half of the volunteers were randomized to start with Treatment A and half were randomized to start with Treatment B
- All doses were taken within 10 minutes after a meal, the order of intake was TMC125 then clarithromycin
- Post-treatment safety visits took place 7 days and 31 (±1) days after the last intake of trial medication
- The study protocol was reviewed and approved by the appropriate institutional ethics committee and health authorities, and was conducted in accordance with the Declaration of Helsinki

Study design

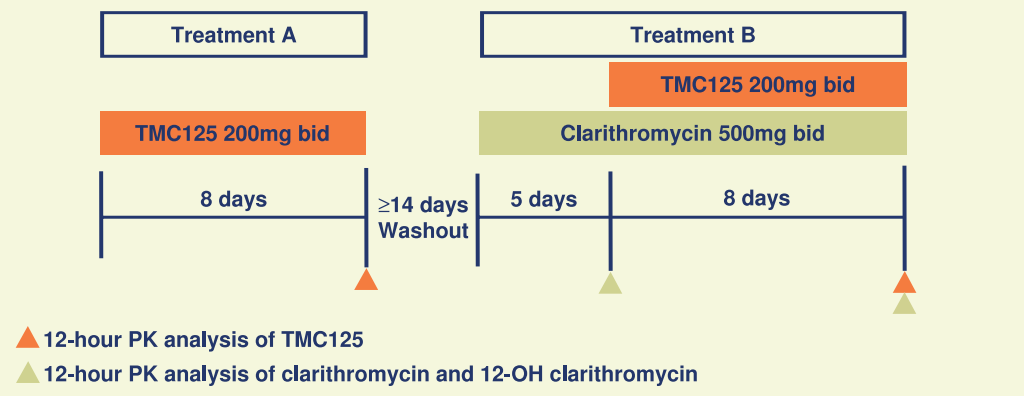
Treatment A
Treatment B
TMC125 200mg bid
TMC125 200mg bid
Clarithromycin 500mg bid
8 days
≥14 days Washout
5 days
8 days
12-hour PK analysis of TMC125
12-hour PK analysis of clarithromycin and 12-OH clarithromycin

- Plasma concentrations of TMC125 were determined for 12 hours on Day 8 of Treatment A and Day 13 of Treatment B
- Plasma concentrations of clarithromycin and 14-OH clarithromycin were determined for 12 hours on Day 5 and Day 13 of Treatment B
- Safety and tolerability assessments were performed throughout the trial until at least 30 days after the last trial medication intake

Methods

- Plasma concentrations of TMC125 were determined using a validated liquid chromatography-tandem mass spectrometry (LC-MS/MS) method (LLOQ 2ng/mL)
- Plasma concentrations of clarithromycin and 14-OH clarithromycin were determined using a validated liquid chromatography method with electrochemical detection (LLOQ 50ng/mL for both compounds)
- PK and statistical PK analyses were performed using
 - WinNonlin Professional™ (version 4.1; Pharsight Corporation, Mountain View, CA, USA)
 - Microsoft Excel® (version 2000; Microsoft, Redmond, WA, USA)
 - SAS (version 9.1.3; SAS Institute Inc., Cary, NC, USA)
- A non-compartmental model with extravascular input was used for the PK analysis

LLOQ = lower limit of quantification

PK and safety parameters and analyses

- Primary PK parameters
 - C_{max} (ng/mL): maximum plasma concentration
 - C_{min} (ng/mL): minimum plasma concentration
 - AUC_{12h} (ng•h/mL): area under the plasma concentration-time curve over a 12-hour period, calculated by linear trapezoidal summation
- Safety parameters
 - Adverse events (AEs), laboratory assessments, ECG, vital signs and physical examinations were evaluated throughout the study
 - severity and drug relationship of AEs to clarithromycin or TMC125 were recorded
- Statistical analyses
 - descriptive statistics were calculated for the PK parameters of TMC125, clarithromycin and 14-OH clarithromycin
 - least square (LS) means were estimated with a linear mixed effects model
 - safety parameters were evaluated by descriptive statistics and frequency tabulations

Demographics

Demographic parameter	All volunteers (n=16)*
Age, years (median[range])	29 (19–49)
Height, cm (median [range])	176 (162–183)
Weight, kg (median [range])	72 (52–89)
BMI, kg/m^2 (median [range])	23 (20–27)
Ethnic origin, n (%)	
Caucasian/White	11 (69)
Black	4 (25)
Hispanic	1 (6)
Gender, n (%)	
Male	16 (100)

*Fifteen volunteers completed the trial (one volunteer withdrew consent)
BMI = body mass index

PK profiles of TMC125

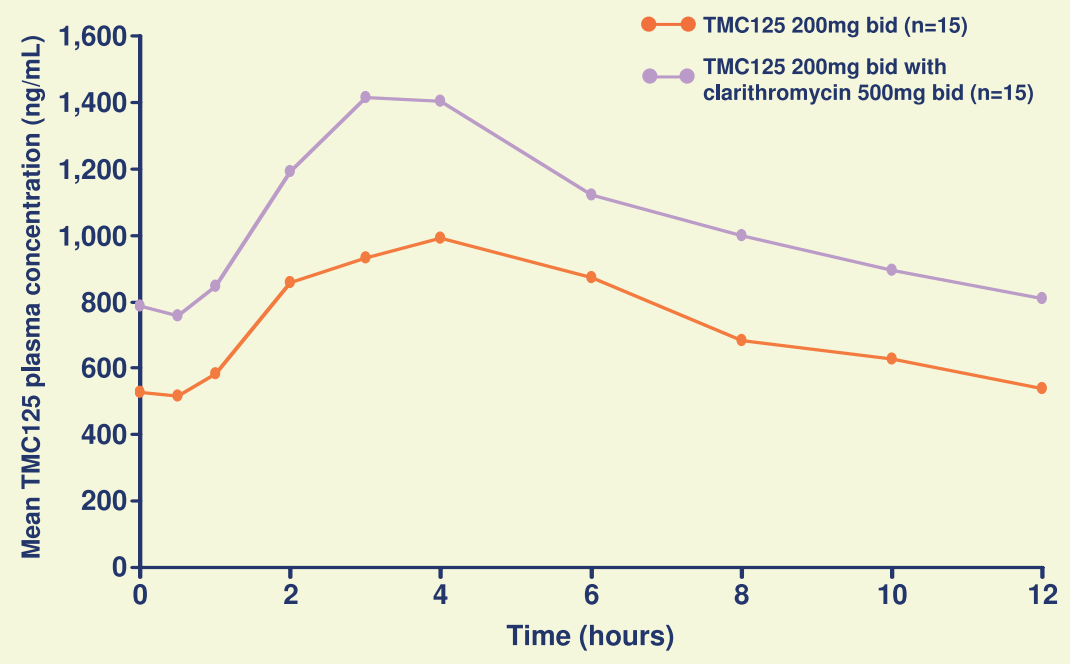
TMC125 200mg bid (n=15)
TMC125 200mg bid with clarithromycin 500mg bid (n=15)
Mean TMC125 plasma concentration (ng/mL)
1,600
1,400
1,200
1,000
800
600
400
200
0
0
2
4
6
8
10
12
Time (hours)

PK profiles of clarithromycin

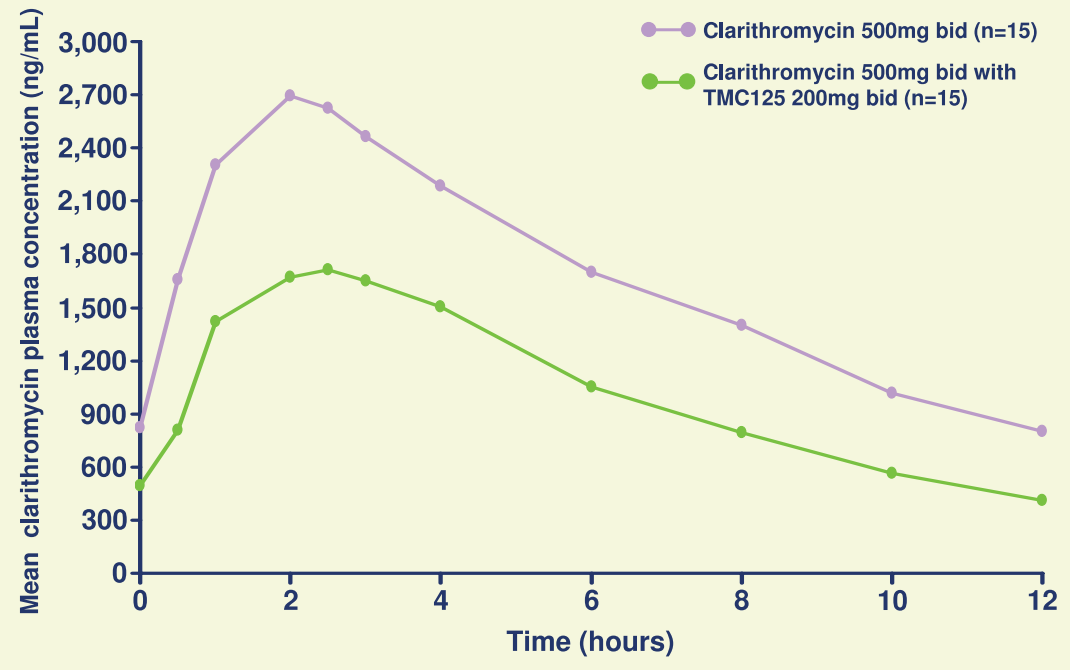
Clarithromycin 500mg bid (n=15)
Clarithromycin 500mg bid with TMC125 200mg bid (n=15)
Mean clarithromycin plasma concentration (ng/mL)
3,000
2,700
2,400
2,100
1,800
1,500
1,200
900
600
300
0
0
2
4
6
8
10
12
Time (hours)

PK profiles of 14-OH clarithromycin

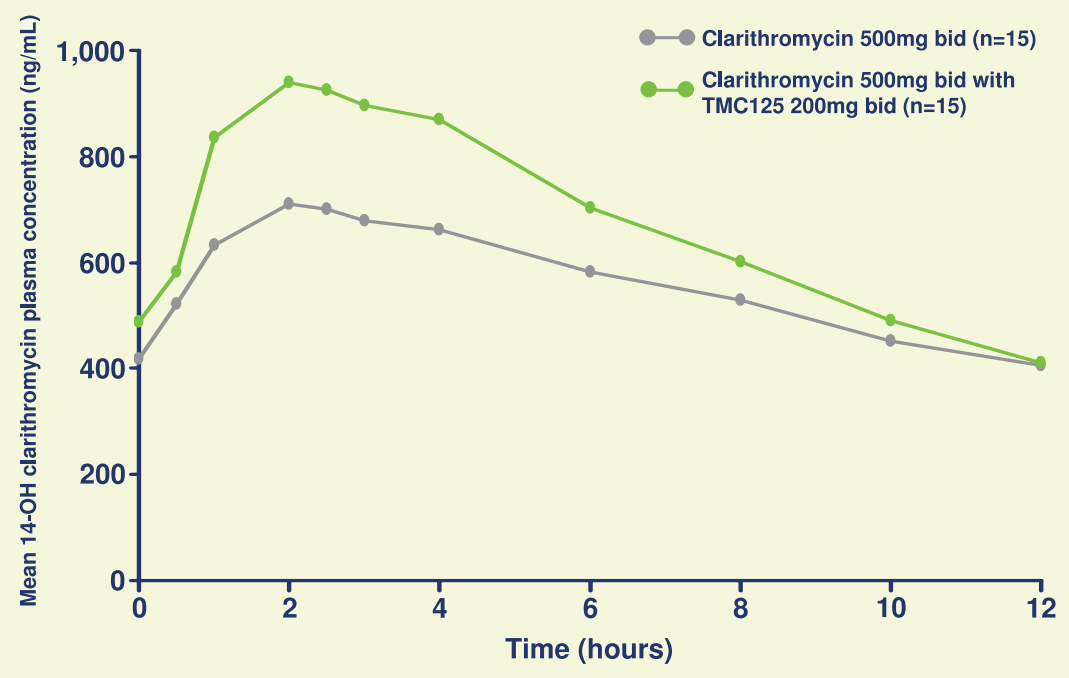
Clarithromycin 500mg bid (n=15)
Clarithromycin 500mg bid with TMC125 200mg bid (n=15)
Mean 14-OH clarithromycin plasma concentration (ng/mL)
1,000
800
600
400
200
0
0
2
4
6
8
10
12
Time (hours)

TMC125 PK parameters (mean ± SD)

PK parameter	TMC125 alone (reference) (n=15)	TMC125 + clarithromycin (test) (n=15)	LS mean ratio (test/reference) (90% CI)
AUC_{12h} (ng•h/mL)	9,008±2,392	12,760±3,559	1.42 (1.34–1.50)
C_{max} (ng/mL)	1,015±244	1,487±390	1.46 (1.38–1.56)
C_{min} (ng/mL)	498±154	726±233	1.46 (1.36–1.58)

SD = standard deviation

Clarithromycin PK parameters (mean ± SD)

PK parameter	Clarithromycin alone (reference) (n=15)	Clarithromycin + TMC125 (test) (n=15)	LS mean ratio (test/reference) (90% CI)
AUC_{12h} (ng•h/mL)	20,240±6,208	12,430±4,248	0.61 (0.53–0.69)
C_{max} (ng/mL)	3,144±917	2,088±572	0.66 (0.57–0.77)
C_{min} (ng/mL)	735±363	371±288	0.47 (0.38–0.57)

14-OH clarithromycin PK parameters (mean ± SD)

PK parameter	Clarithromycin alone (reference) (n=15)	Clarithromycin + TMC125 (test) (n=15)	LS mean ratio (test/reference) (90% CI)
AUC_{12h} (ng•h/mL)	6,761±1,893	8,183±2,100	1.21 (1.05–1.39)
C_{max} (ng/mL)	766±205	1,030±318	1.33 (1.13–1.56)
C_{min} (ng/mL)	382±134	394±109	1.05 (0.90–1.22)

Safety summary

- No serious AEs, or grade 3 or 4 AEs were reported
- No volunteer discontinued the trial due to an AE
- All AEs were mild in severity except one case of grade 2 abdominal pain, not related to the trial medication
- No cases of rash were reported
- No consistent or relevant changes were found in laboratory or cardiovascular safety parameters, or physical examinations

Conclusions

- When co-administered with clarithromycin 500mg bid, TMC125 exposure was increased by 42% with similar increases in C_{max} and C_{min}, probably due to inhibition of CYP450 enzymes by clarithromycin. This increase of exposure is not believed to be clinically relevant given the safety profile of TMC125 and that no PK/safety relationship has so far been observed for TMC125 in clinical studies.
- When co-administered with TMC125, clarithromycin exposure was decreased by 39% and exposure to its metabolite 14-OH clarithromycin was increased by 21%, probably due to the induction of CYP3A4 by TMC125.
- Short-term co-administration of TMC125 with clarithromycin in HIV-negative volunteers was generally safe and well tolerated.
- No dose adjustment of TMC125 is recommended.
- Since 14-OH clarithromycin has reduced activity against MAC, overall activity against this pathogen may be decreased when combined with TMC125. Therefore alternatives to clarithromycin, such as azithromycin, should be considered for this infection.

References

1. Vingerhoets J, et al. J Virol 2005;79:12773–82.
2. Nadler J, et al. 10th European AIDS Conference 2005 (Poster LBPS3/7A).
3. Cohen C, et al. 16th International AIDS Conference 2006 (Poster TUPE0061).
4. Rodvold KA. Clin Pharmacokinet 1999;37:385–98.

Acknowledgements

The authors would like to express their gratitude to the volunteers. We also acknowledge:

- MP Bouche, J&J Pharmaceutical Research and Development, Beerse, Belgium
- T Duvauchelle, Aster Research Unit, Paris, France.

Presented at the 44th Annual Meeting of the Infectious Diseases Society of America, October 12–15, 2006, Toronto, Canada. This poster is available on-line at www.tibotec.com.